LB SUN 71

A Phase IIa Study of BGS649, an Oral Aromatase Inhibitor for the Treatment of Men with Obesity-Associated Secondary Hypogonadotropic Hypogonadism

Mohamed Bidair[1], Jackie Parkin[2], Alastair MacKinnon[2]

[1]San Diego Clinical Trials, San Diego, CA, USA; [2]Mereo BioPharma Group plc, London, UK

Mereo BioPharma

Background

- Aromatase is highly expressed in adipose tissue, where it converts testosterone to estradiol[1]
 - Excess adipose tissue is associated with excess aromatase activity in obese patients; this results in high estradiol levels[2]
 - Excess estradiol can feed back to the hypothalamic pituitary axis (HPA), suppressing gonadotropin secretion, testicular testosterone production and spermatogenesis in obese men[2]
- Thus, severe obesity in men is associated with symptomatic androgen deficiency, a condition known as obesity-associated hypogonadotropic hypogonadism (OHH)
- Current treatment options for OHH are limited:
 - Testosterone replacement therapies carry risks of supra-physiological testosterone levels, infertility as a result of follicle stimulating hormone (FSH)/ luteinizing hormone (LH) suppression
 - Topical administration of testosterone can cause transference to partners and children
- BGS649 is an oral, once-weekly aromatase inhibitor, which is under investigation as a treatment for OHH:
 - Designed to be used in low-dose regimens suitable for endocrine indications with a long half-life enabling convenience of once-weekly dosing
 - Mechanism of action anticipated to restore testosterone whilst maintaining HPA feedback, thereby avoiding risks of supra-physiological testosterone levels while maintaining gonadotrophins and testicular function

Study objective

- This was a proof-of-concept, dose-finding, two-part study of efficacy and safety/tolerability of BGS649 for the treatment of men with obesity-associated secondary OHH:
 - Specifically to determine doses of BGS649 that increase and maintain total testosterone levels within the normal range (Part 1)
 - To assess the impact of BGS649 normalization of testosterone on metabolic clinical endpoints (Part 2)
 - To explore the safety and tolerability of BGS649 in the OHH population, including in a randomized, placebo-controlled study (Part 2)

Methods

Study design

- This Phase II study comprised two separate parts:
 - Part 1: a 12-week, non-randomized, open-label, BGS649 dose-finding study
 - Part 2: a 12-week, randomized, double-blind study of BGS649 versus placebo
- ClinicalTrials.gov: NCT01200862

Patients

- Adult males with body mass index (BMI) ≥30 kg/m^2
- Morning testosterone levels (<300 ng/dL) on ≥2 separate occasions
- Estradiol levels within or above normal range
- Inappropriately low gonadotropins consistent with secondary hypogonadism
 - LH levels below normal range
 - FSH levels below normal range
- No underlying endocrinopathy/pituitary disease

Figure 1: Study design

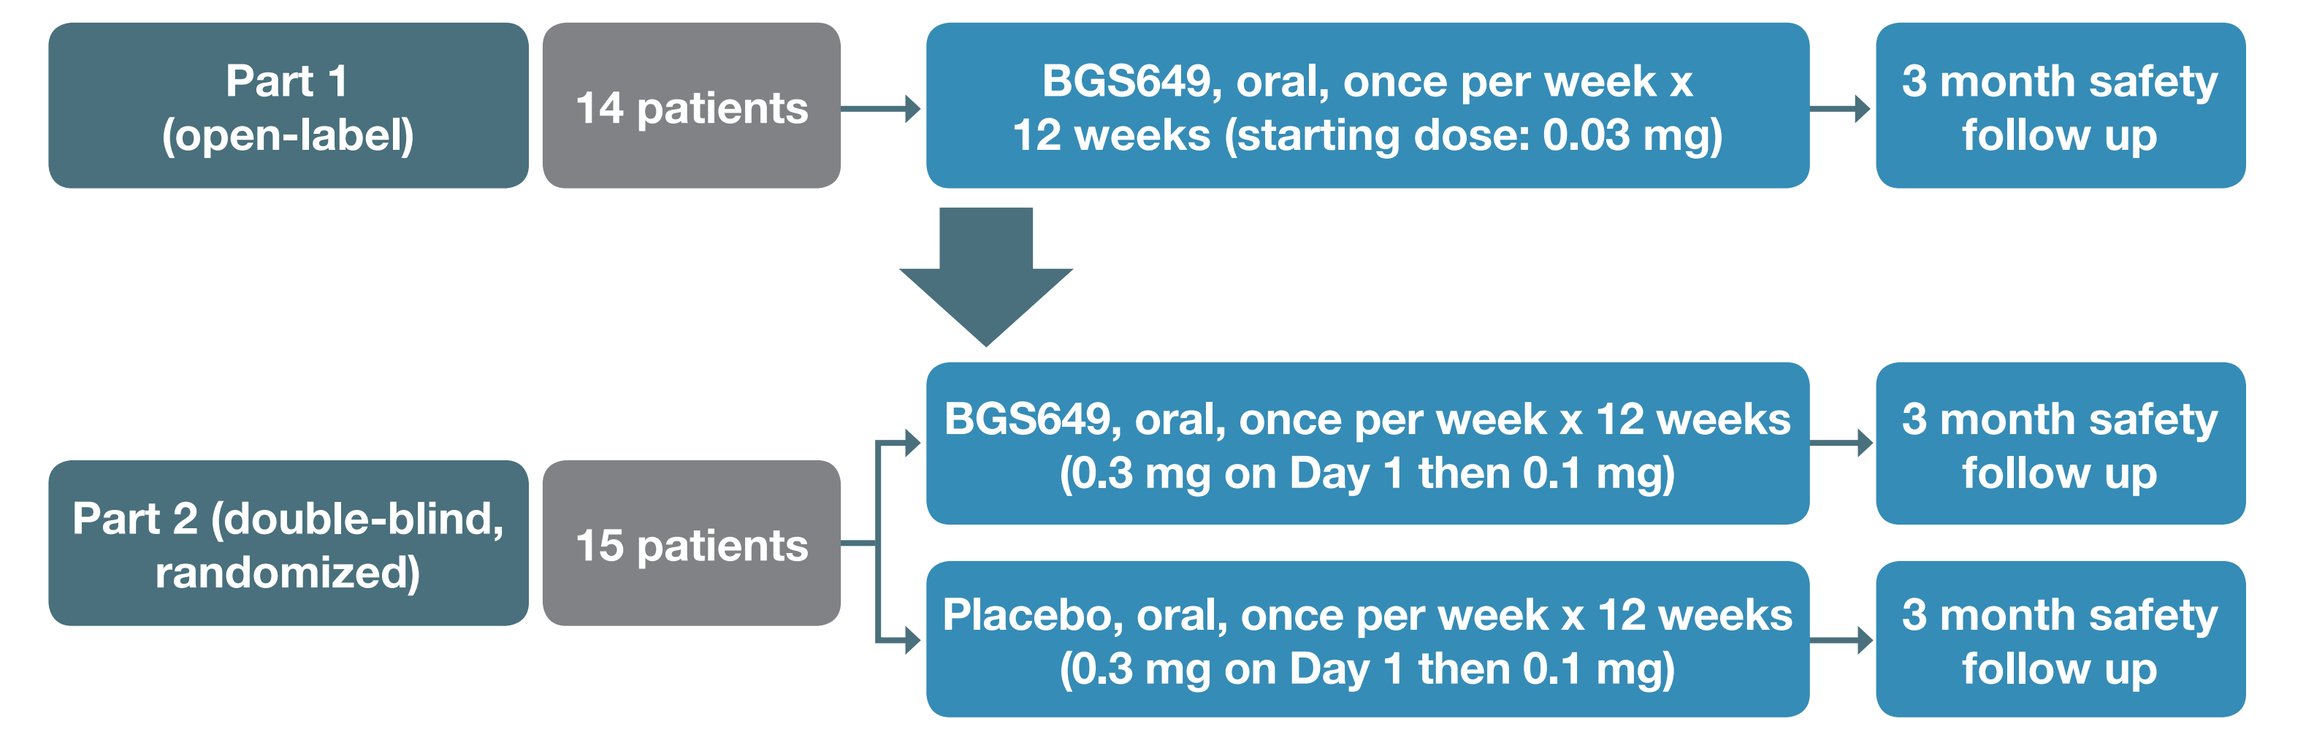

Part 1
(open-label)
14 patients
BGS649, oral, once per week x
12 weeks (starting dose: 0.03 mg)
3 month safety
follow up
Part 2 (double-blind,
randomized)
15 patients
BGS649, oral, once per week x 12 weeks
(0.3 mg on Day 1 then 0.1 mg)
3 month safety
follow up
Placebo, oral, once per week x 12 weeks
(0.3 mg on Day 1 then 0.1 mg)
3 month safety
follow up

Treatment

- Part 1: starting dose was 0.03 mg BGS649, titrated weekly until total and free testosterone were within normal levels (maximum allowable single dose 5.0 mg)
- Part 2: BGS649 dosing was determined by Part-1 pharmacokinetic (PK) / pharmacodynamic (PD) analyses and was 0.3 mg Day 1 and 0.1 mg at all subsequent visits (total of 11 weekly doses)

Primary endpoint:

- **Part 1:** total testosterone levels
- **Part 2*:** insulin sensitivity by:
 - Homeostatic Model Assessment of Insulin Resistance (HOMA-IR)
 - Quantitative Insulin-sensitivity Check Index (QUICKI)

Secondary endpoints (both parts):

- Testosterone normalization
- Gonadotropins FSH and LH
- Estradiol
- Metabolic parameters: glycated hemoglobin (HbA1c), glucose tolerance, lipids, insulin sensitivity
- Safety/tolerability: adverse events (AEs); hematology, chemistry and urinalysis; semen parameters; bone turnover biomarkers and bone mineral density (BMD)

Part 2 was stopped early when proof of concept for testosterone normalization was observed and re-evaluation of the sample size showed the study was underpowered for insulin sensitivity analyses; this (along with the discovery of dosing errors in the placebo arm) contributed to the decision for early termination of Part 2 of the study

Patient demographics

- **Part 1:** 14 patients were enrolled and 13 completed the study (1 patient was lost to follow-up)
- **Part 2:** 15 patients were enrolled, but not all completed the study due to early study termination outlined above:
 - 7 patients received BGS649
 - 5 patients completed 12 weeks' treatment
 - 1 patient completed 8 weeks' dosing and 1 patient completed 6 weeks' dosing
 - 8 patients received placebo
 - 3 patients completing 12 weeks' treatment
 - 2 patients completed to Week 10, 1 patient to Week 7, 1 patient to Week 6 and 1 patient to Week 2
- Baseline characteristics were similar in the two parts of the study:
 - **Part 1:** Male patients with a mean age of 50.3 years, and mean BMI of 33.952 kg/m^2; 12 (86%) patients were Caucasian and 2 (14%) were Black
 - **Part 2:** Male patients with mean age of 50.3 years, and mean BMI of 37.868 kg/m; 13 (86%) patients were Caucasian, 1 (7%) was Black and 1 (7%) was Native American

Figure 2: Primary endpoint Part 1: Mean (±SE) profiles of percent change from baseline for total testosterone

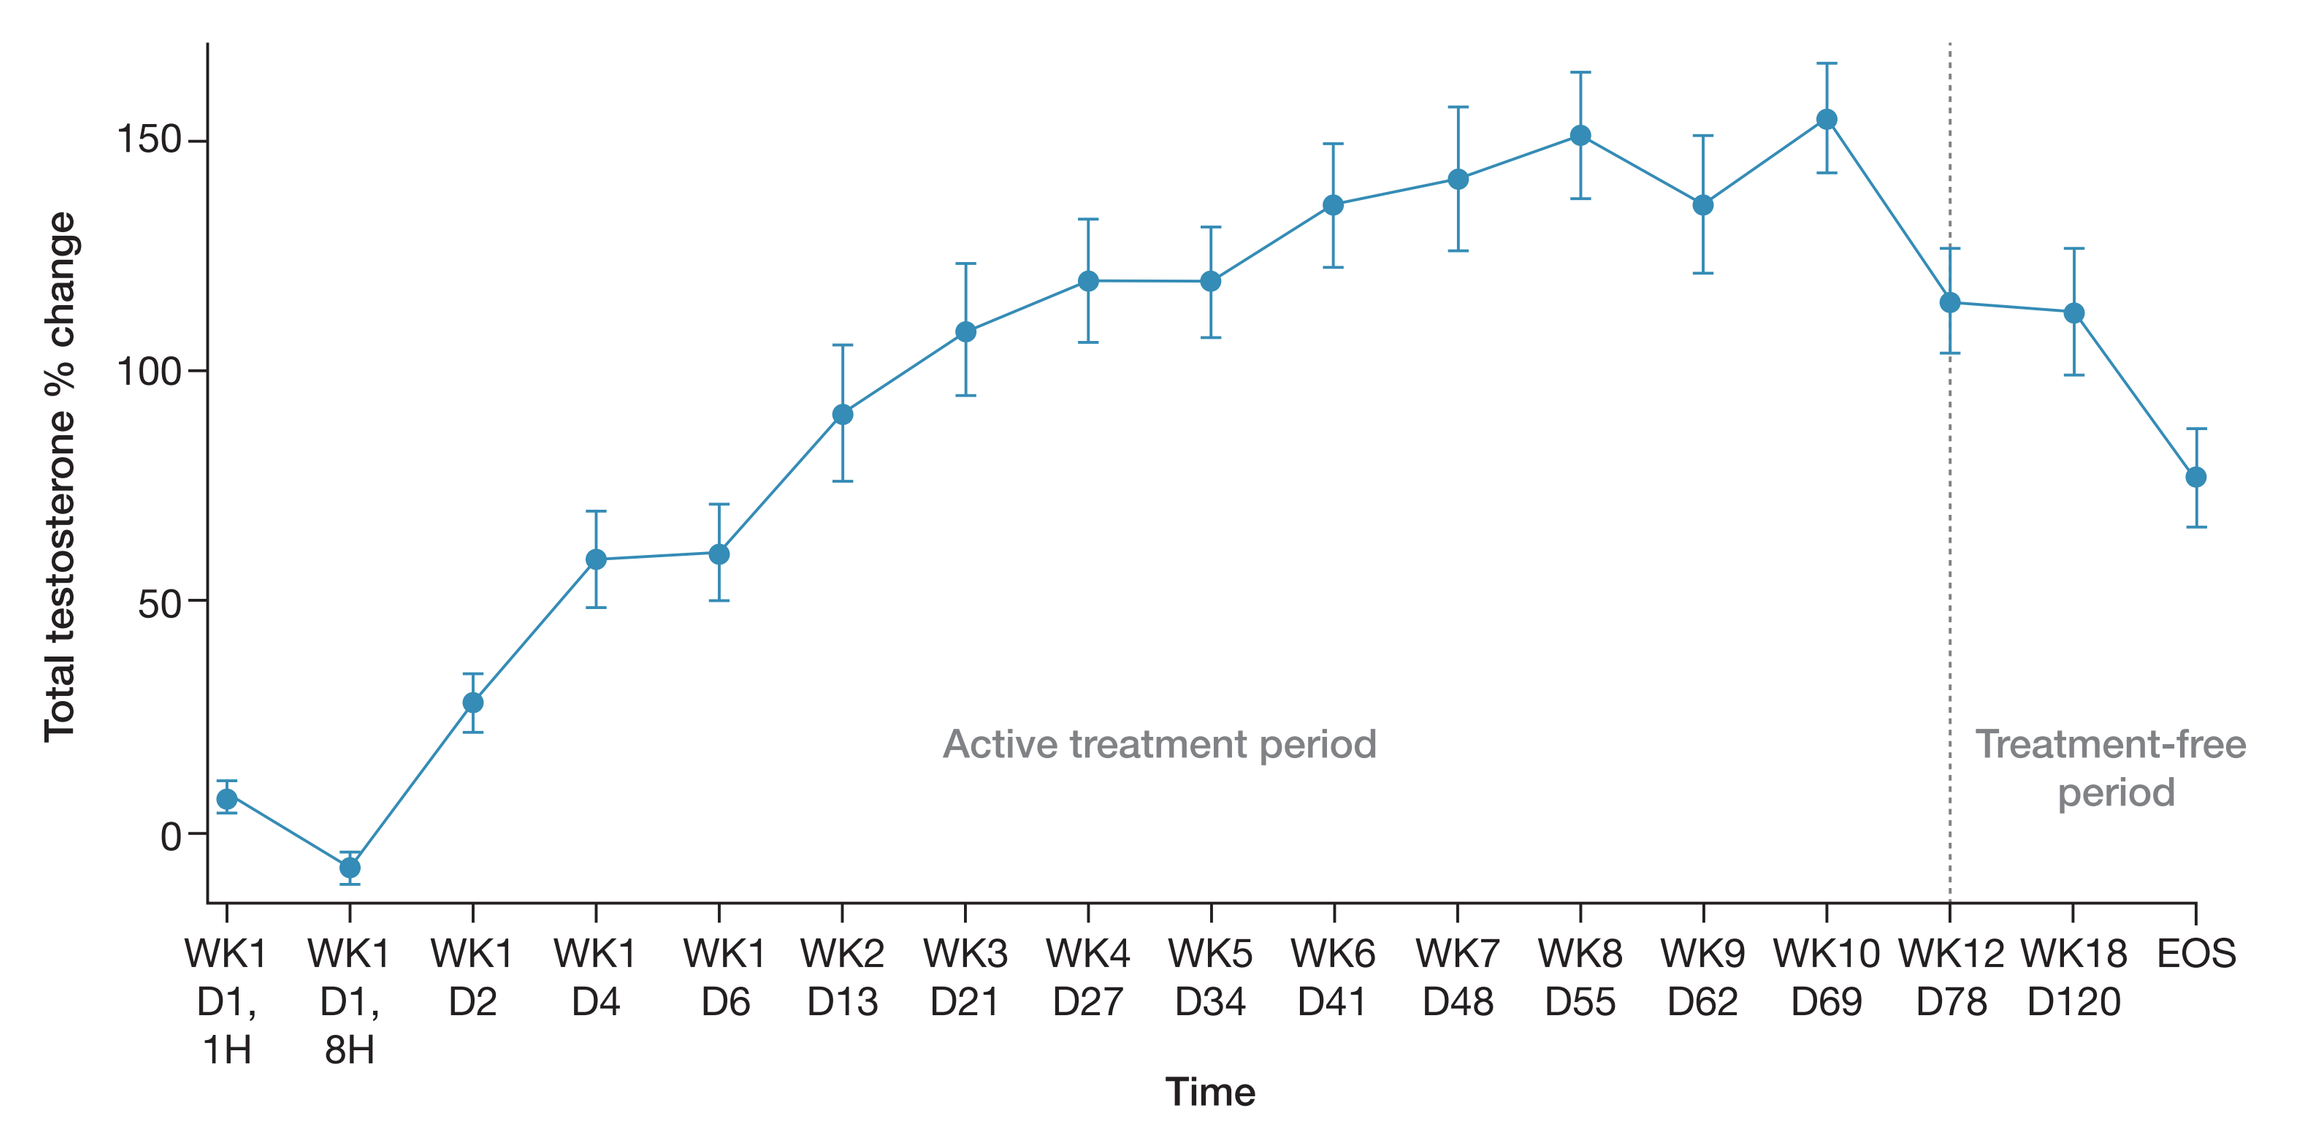

Total testosterone % change
150
100
50
0
Active treatment period
Treatment-free period
WK1 D1, 1H
WK1 D1, 8H
WK1 D2
WK1 D4
WK1 D6
WK2 D13
WK3 D21
WK4 D27
WK5 D34
WK6 D41
WK7 D48
WK8 D55
WK9 D62
WK10 D69
WK12 D78
WK18 D120
EOS
Time

BAS, baseline, EOS, end of study; SCR, screening; SE, standard error

Efficacy results: Part 1

Total testosterone levels (primary endpoint)

- Dose titration with BGS649 increased total testosterone levels from baseline to Week 12 to within normal levels (mean ± SD: 239 ± 52 to 514 ± 188 ng/dL [n=14])
- Increases in testosterone were rapid, seen at Week 1, with all 14 patients reaching normal levels by Week 4, and 13/13 patients maintaining normal levels through Week 12 (one patient was lost to follow-up)
- There was no testosterone elevation above the normal range

Other sex hormones (estradiol, FSH and LH)

- FSH increased by 135% and LH increased by 100% from baseline to Week 12 consistent with the increases in testosterone levels with BGS649
- There was a 30% reduction from baseline to Week 12 in estradiol levels with BGS649 treatment reflecting the mechanism of action

Table 1: Hormone levels at baseline and Week 12

Parameter, mean ± SD	N	Baseline	N	Week 12
Total testosterone, ng/mL	14	239 ± 52	14	514 ± 188
Estradiol, ng/mL	11	26.2 ± 7.2	11	18.2 ± 9.6
FSH, IU/L	14	4.06 ± 1.62	14	9.56 ± 6.03
LH, IU/L	14	6.05 ± 2.59	14	12.12 ± 4.77

FSH, follicle stimulating hormone; IU, international unit; LH, luteinizing hormone; SD, standard deviation

Efficacy results: Part 2

Testosterone normalization

- Treatment with BGS649 resulted in normalization of baseline testosterone levels from baseline to Week 12 to within normal levels in all patients; levels returned towards baseline by EOS (mean ± SD: 273 ± 142 to 423 ± 70 ng/dL [n=5])
- Increases in testosterone levels occurred quickly and were seen as early as Week 1
- Placebo did not show an elevation in testosterone levels from baseline to Week 12 (mean: 309 to 366 ng/dL [n=1])
- There was no testosterone elevation above the normal range

Insulin sensitivity (primary endpoint)

- Based on Part-1 PK/PD analyses, BGS649 was dosed in Part 2 at 0.3 mg (Week 1), then 0.1 mg (Weeks 2–11) to maintain drug plasma levels above the IC90 for aromatase inhibition
- Data for insulin sensitivity were therefore deemed insufficient to allow meaningful interpretation. BGS649 had no effect on other metabolic parameters (HbA1c, glucose tolerance, or lipids)
- Testosterone normalization was further confirmed versus placebo following 12 weeks of treatment (Part 2)

Figure 3: Part 2: Mean (±SE) profiles of percent change from baseline for total testosterone

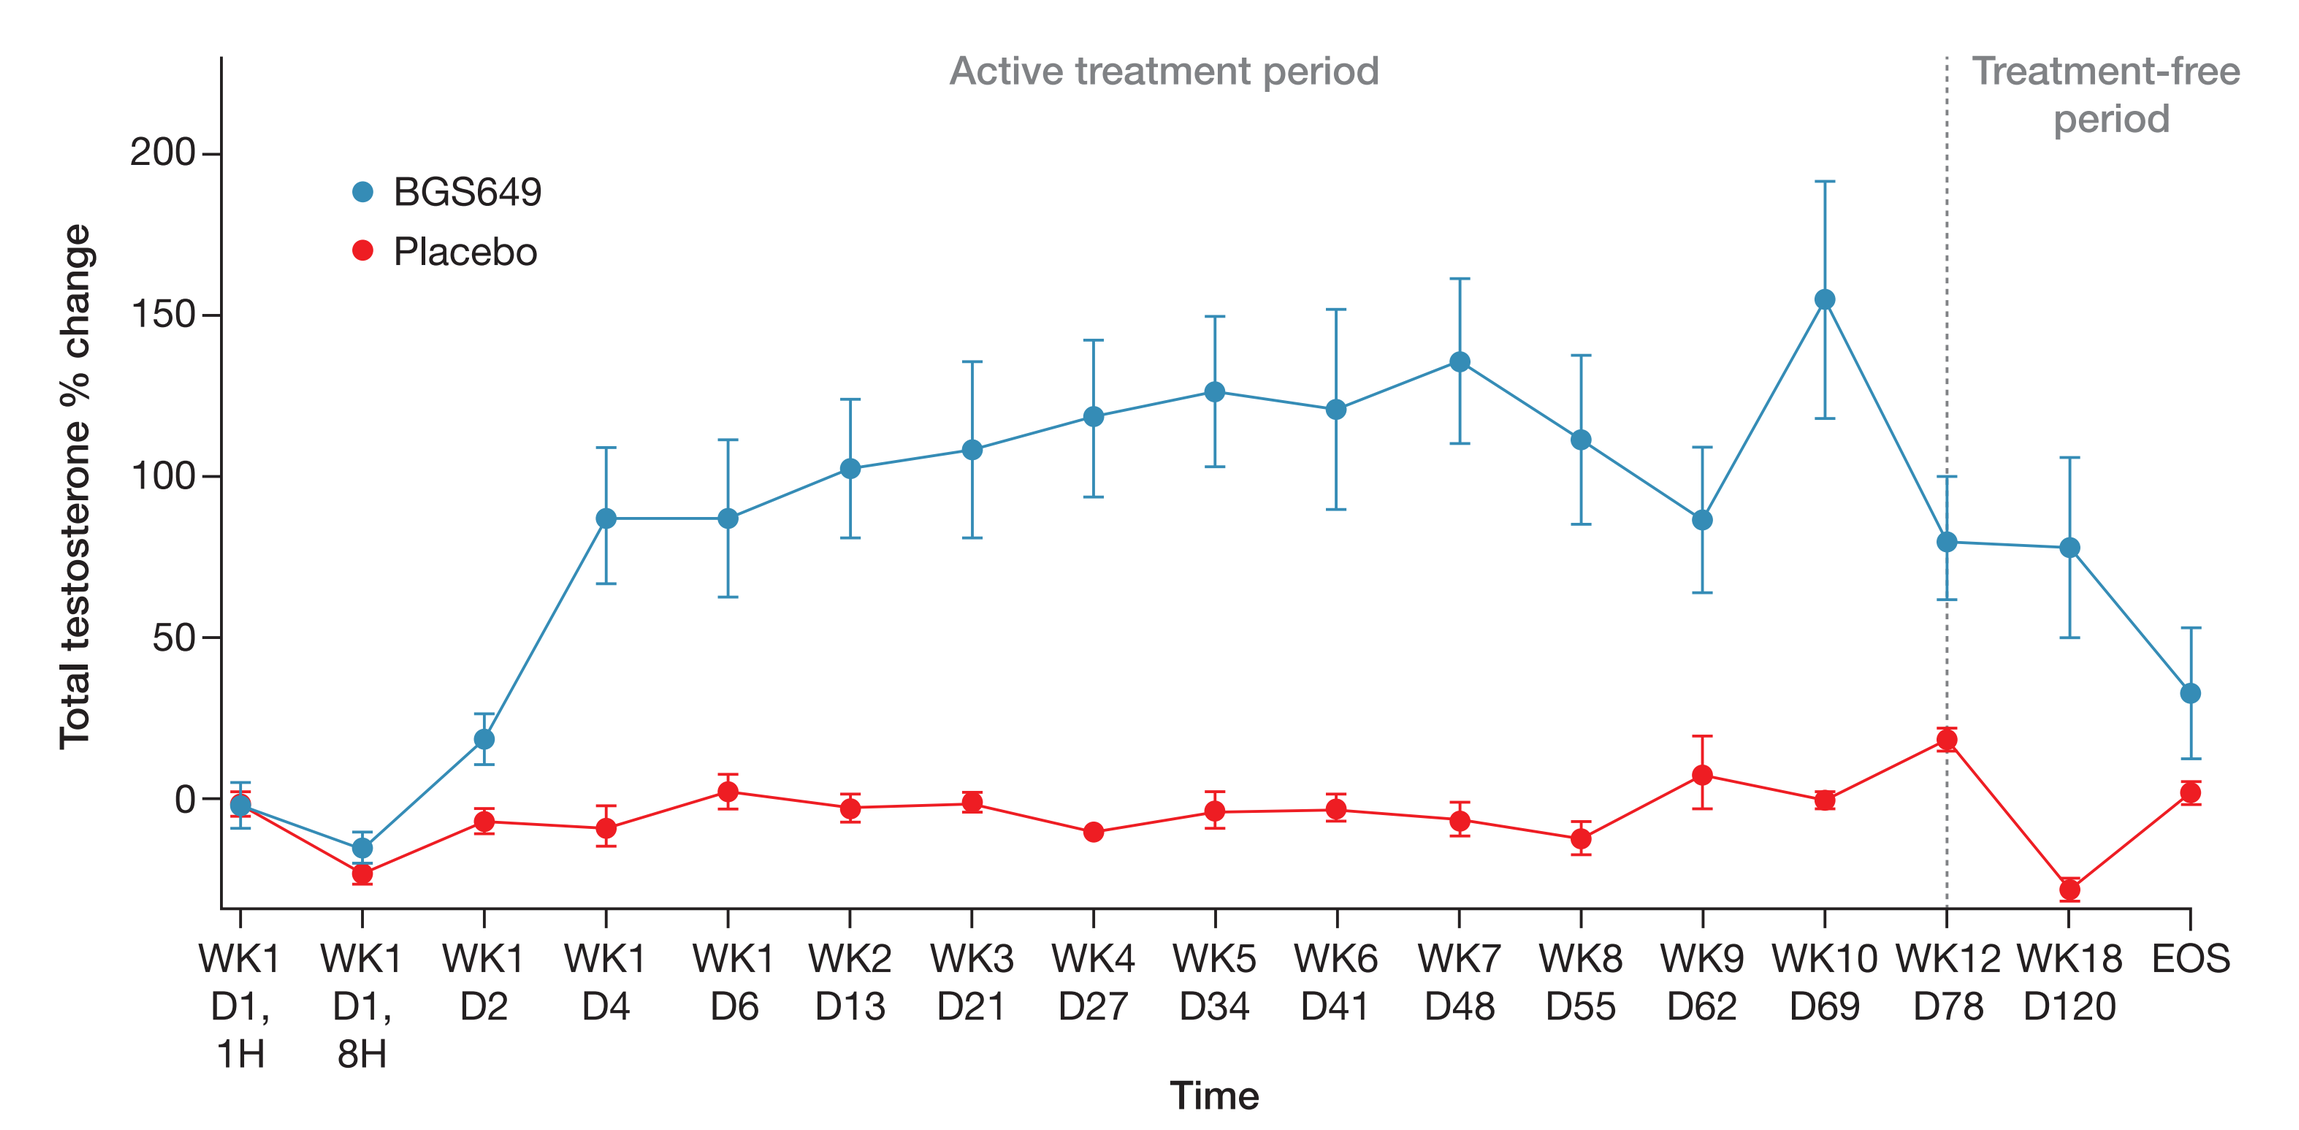

Active treatment period
Treatment-free period
BGS649
Placebo
Total testosterone % change
200
150
100
50
0
WK1 D1, 1H
WK1 D1, 8H
WK1 D2
WK1 D4
WK1 D6
WK2 D13
WK3 D21
WK4 D27
WK5 D34
WK6 D41
WK7 D48
WK8 D55
WK9 D62
WK10 D69
WK12 D78
WK18 D120
EOS
Time

BAS, baseline, EOS, end of study; SCR, screening; SE, standard error

Total testosterone levels

- Normal testosterone levels were sustained with BGS649 from baseline to Week 12, but not with placebo
- Increases in testosterone levels were seen as early as Week 1 and normalized by EOS
- There was no testosterone elevation above the normal range

Other sex hormones (estradiol, FSH and LH)

- Estradiol levels in patients receiving BGS649 were reduced compared with those in patients receiving placebo over the treatment period
- FSH and LH levels increased in patients treated with BGS649 compared with those in patients receiving to placebo over 12 weeks

Table 2: Hormone levels at baseline, Week 10 and Week 12 for BGS649 and placebo (per-protocol)

Parameter, mean ± SD	BGS649						Placebo					
	N	Baseline	N	Week 10	N	Week 12	N	Baseline	N	Week 10	N	Week 12
Total testosterone, ng/dL	5	273 ± 142	5	569 ± 129	5	423 ± 70	3	256 ± 51	3	257 ± 63	1	366
Estradiol, ng/dL	5	31.0 ± 15.4	5	26.4 ± 14.0	5	14.0 ± 2.1	3	17.3 ± 4.2	3	28.7 ± 12.9	1	46.0
FSH, IU/L	3	6.70 ± 4.03	3	9.07 ± 4.30	3	9.40 ± 4.21	2	6.75 ± 3.89	2	6.80 ± 2.69	1	2.60
LH, IU/L	3	5.97 ± 4.20	3	5.70 ± 2.70	3	9.52 ± 4.40	2	3.05 ± 0.35	2	6.20 ± 2.40	1	4.50

FSH, follicle stimulating hormone; IU, international unit; LH, luteinizing hormone; SD, standard deviation

Safety/Tolerability results (Part 1 and Part 2)

Adverse events

- There were no deaths in the study, no discontinuations due to AEs related to study drug, and no SAEs on BGS649 treatment
- **Part 1**: 11 patients had a total of 59 AEs, the most common of which were: spontaneous penile erection, headache, oropharyngeal pain, nasal congestion, diarrhoea, arthralgia, cough, dizziness, frequent bowel movements, and abnormal hair growth
- **Part 2**: 5 patients from the BGS649 group had a total of 16 AEs and 7 patients from the placebo group had a total of 25 AEs, the most common of which were: headache, asthenia, increased appetite, nasal congestion, somnolence, and spontaneous penile erection
- All AEs in BGS649 arms were mild or moderate, transient and resolved without intervention

Figure 4: Part 2: Mean ± profiles of percent change from baseline in FSH

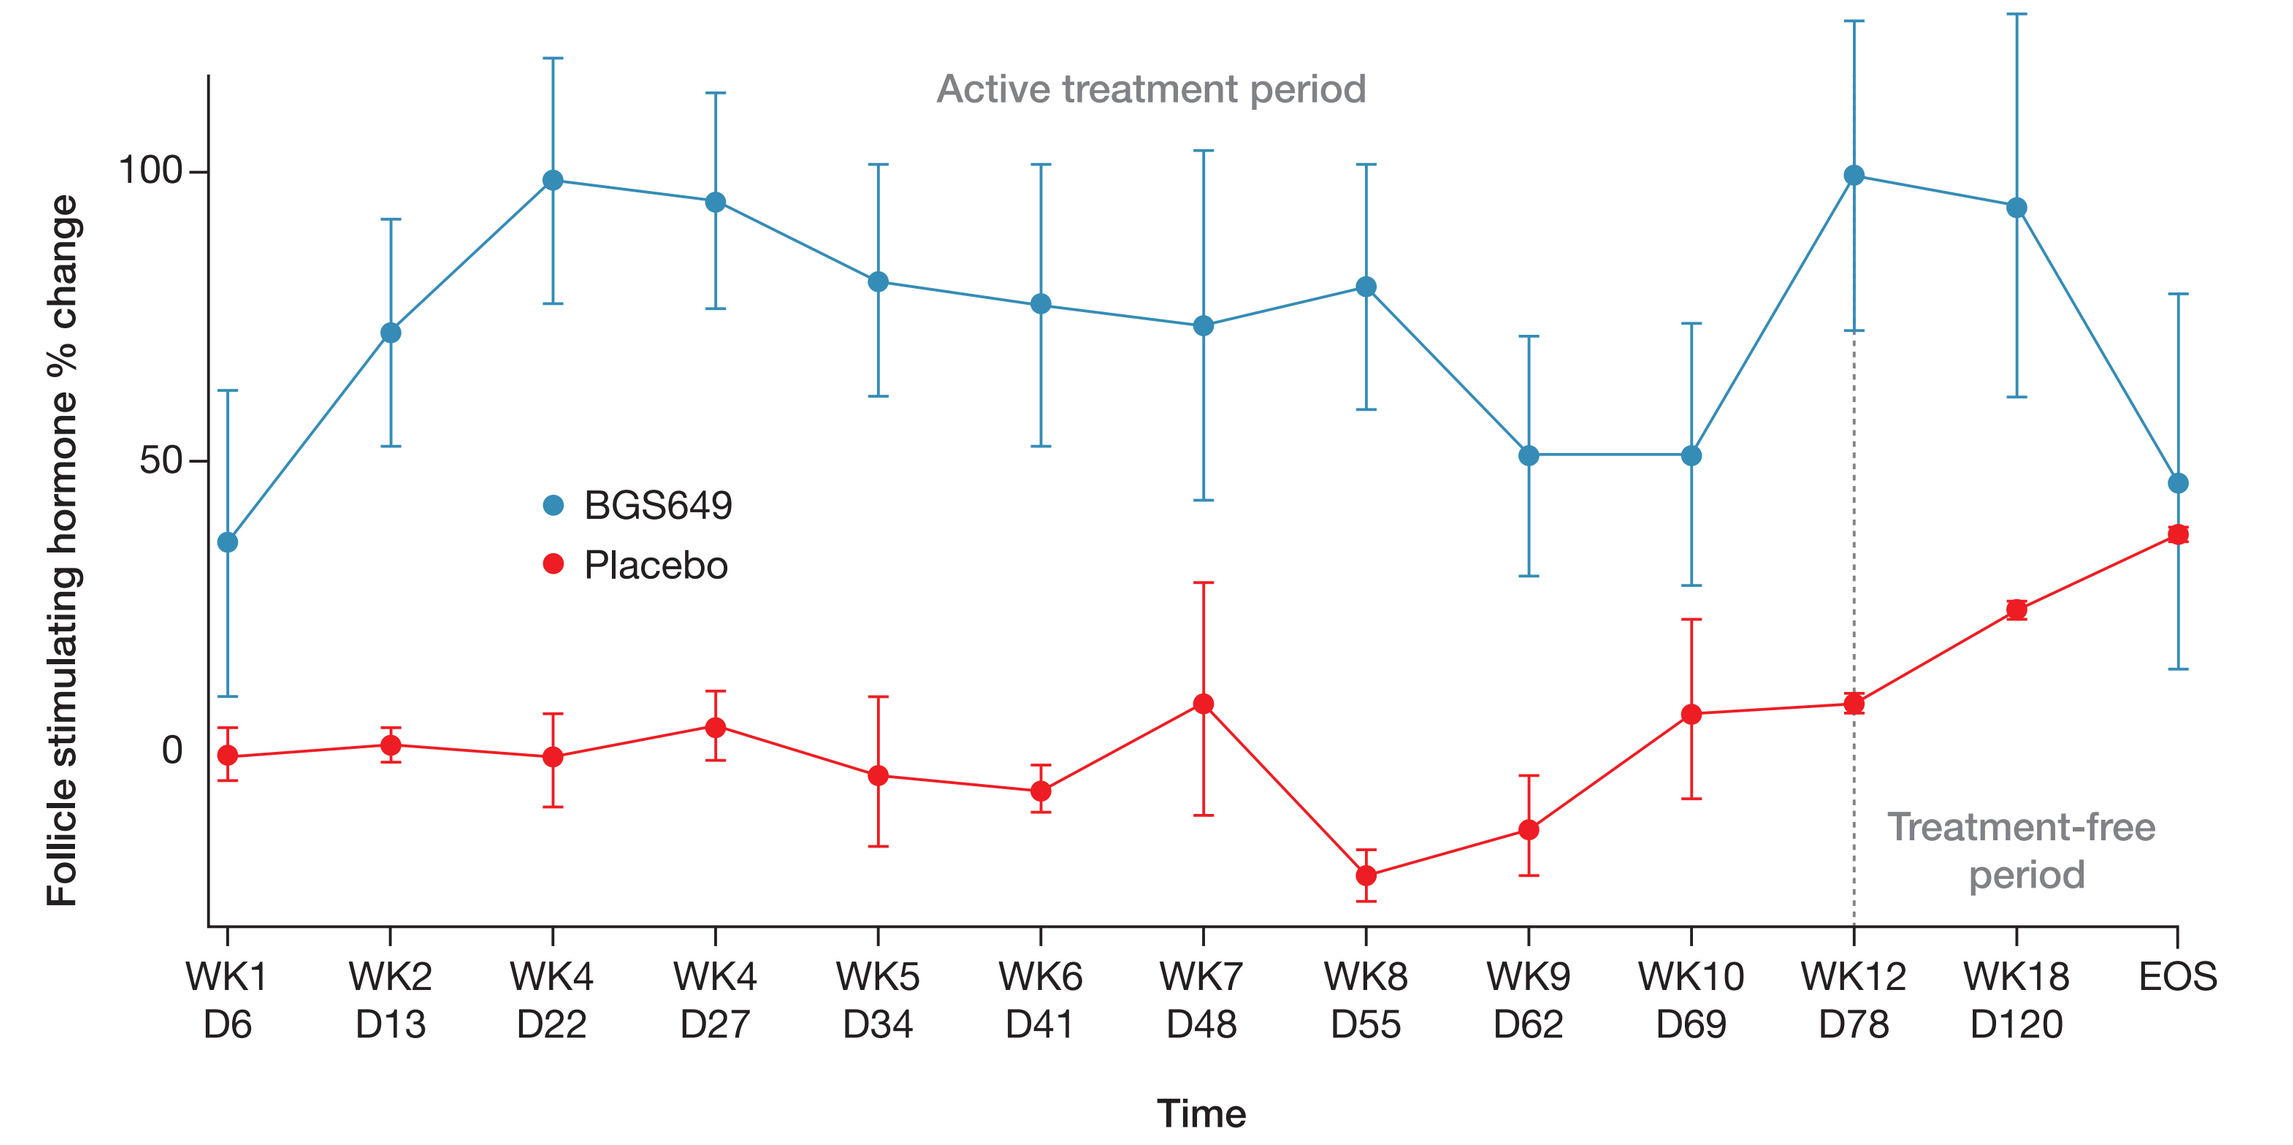

Active treatment period
Treatment-free period
BGS649
Placebo
Follicle stimulating hormone % change
100
50
0
WK1 D6
WK2 D13
WK4 D22
WK4 D27
WK5 D34
WK6 D41
WK7 D48
WK8 D55
WK9 D62
WK10 D69
WK12 D78
WK18 D120
EOS
Time

EOS, end of study; SCR, screening; BAS, baseline

Table 3: Most common treatment-emergent adverse events (those in 2 patients or more in either treatment or placebo groups)

AE, n (%)	Part 1	Part 2	
	BGS649 (n=14)	BGS649 (n=7)	Placebo (n=8)
Any AE	11 (78.6)	5 (71.4)	7 (87.5)
Headache	4 (28.6)	2 (28.6)	1 (12.5)
Spontaneous penile erection	4 (28.6)	1 (14.3)	1 (12.5)
Diarrhoea	3 (21.4)	0 (0)	0 (0)
Nasal congestion	3 (21.4)	1 (14.3)	1 (12.5)
Oropharyngeal pain	3 (21.4)	1 (14.3)	0 (0)
Arthralgia	2 (14.3)	0 (0)	0 (0)
Cough	2 (14.3)	1 (14.3)	0 (0)
Dizziness	2 (14.3)	0 (0)	0 (0)
Frequent bowel movements	2 (14.3)	0 (0)	0 (0)
Abnormal hair growth	2 (14.3)	0 (0)	0 (0)
Asthenia	0 (0)	0 (0)	2 (25.0)
Somnolence	0 (0)	0 (0)	2 (25.0)

Data are for adverse events in treatment period only, and exclude treatment free follow-up period

Laboratory parameters

- There were no clinically significant hematology, urinalysis or clinical chemistry abnormalities, outside of a single case of elevated triglycerides and no changes in semen parameters
- In particular, there were no significant changes in hemoglobin, hematocrit and prostate specific antigen
- There was no effect on bone turnover biomarkers (BSAP, CTx, P1NP and osteocalcin) or bone mineral density

Conclusions

- BGS649 is an oral aromatase inhibitor specifically designed for low-dose, once-weekly use and in development for the treatment of men with OHH
- Once-weekly BGS649 restored normal physiological testosterone levels maintained over 12 weeks in men with OHH, the effects being observed as early as 1 week with no testosterone overshoot
- Increases in both FSH and LH were seen with BGS649 across the 12 weeks consistent with its mechanism of action; these positive effects support the potential for BGS649 to maintain or improve testicular function
- Decreases in estradiol were observed consistent with BGS649 mode of action
- BGS649 was generally safe and well-tolerated with no severe AEs or drug-related study discontinuations during the entire study period
- To further evaluate the testosterone normalization, clinical benefit and safety of BGS649, a larger 6-month dose-ranging Phase IIb study of BGS649 in men with OHH is ongoing (NCT02730169) with a 12-month extension study (NCT02908074)

References

1. Stocco C. *Steroids* 2012;77:27-35.
2. Pitteloud N et al. *J Clin Endocrinol Metab* 2008;93:2686-2692.

Acknowledgements

We thank all of the study investigators and participants

We thank Nick Brown of iS LifeScience for providing medical writing support funded by Mereo BioPharma

This study was originally sponsored by Novartis; this investigational medicinal product is now under clinical development sponsored by Mereo BioPharma, who are carrying out all ongoing clinical studies

Disclosures

Mohamed Bidair has no disclosures

Jackie Parkin & Alastair MacKinnon are employees of Mereo BioPharma

Presented at The Endocrine Society's 99th Annual Meeting; Orlando, FL, USA; April 1-4, 2017

www.mereobiopharma.com