111P

Interim biomarker analysis of a Phase 1b/2 Study of anti-TIGIT Etigilimab (MPH313) and Nivolumab in Subjects with Select Locally Advanced or Metastatic Solid Tumors (ACTIVATE)

Ghanashyam Sarikonda[1], Breanna K. Wallace[1], Cedric Wiesner[2], Suba Krishnan[1], John Lewicki[1] and Ann M. Kapoun[1]
[1]Mereo Biopharma, Mountain View, CA, USA [2]Wiesner Consulting LLC, Livermore, CA, USA

INTRODUCTION

T cell immunoreceptor with immunoglobulin and immunoreceptor tyrosine-based inhibitory motif domain (TIGIT) is a co-inhibitory receptor of T cell and natural killer cell activity. Etigilimab (etig), is an FCγR competent, humanized anti-TIGIT IgG1 monoclonal antibody that blocks its interaction with PVR (poliovirus receptor) and inhibits downstream signaling. As co-targeting TIGIT in combination with anti-PD-1 could enhance anti-tumor immunity, biomarker effects of this combination are being studied in ACTIVATE, an open-label Phase 1b/2 basket study evaluating efficacy, safety, tolerability and PK/PD of etig+nivo.

Biomarker monitoring was included as an exploratory endpoint following etig+Nivo combination treatment. Peripheral blood (PB) samples were evaluated for activation of immunological modulators by flow cytometry. Plasma samples were evaluated for changes in cytokines and circulating tumor DNA (ctDNA). Tissue (FFPE) samples were analyzed at baseline to determine expression of various immune parameters including PD-L1 by cIHC or multiplex Immuno-fluorescence and mRNAseq.

RESULTS

TIGIT and PVR expression by IHC

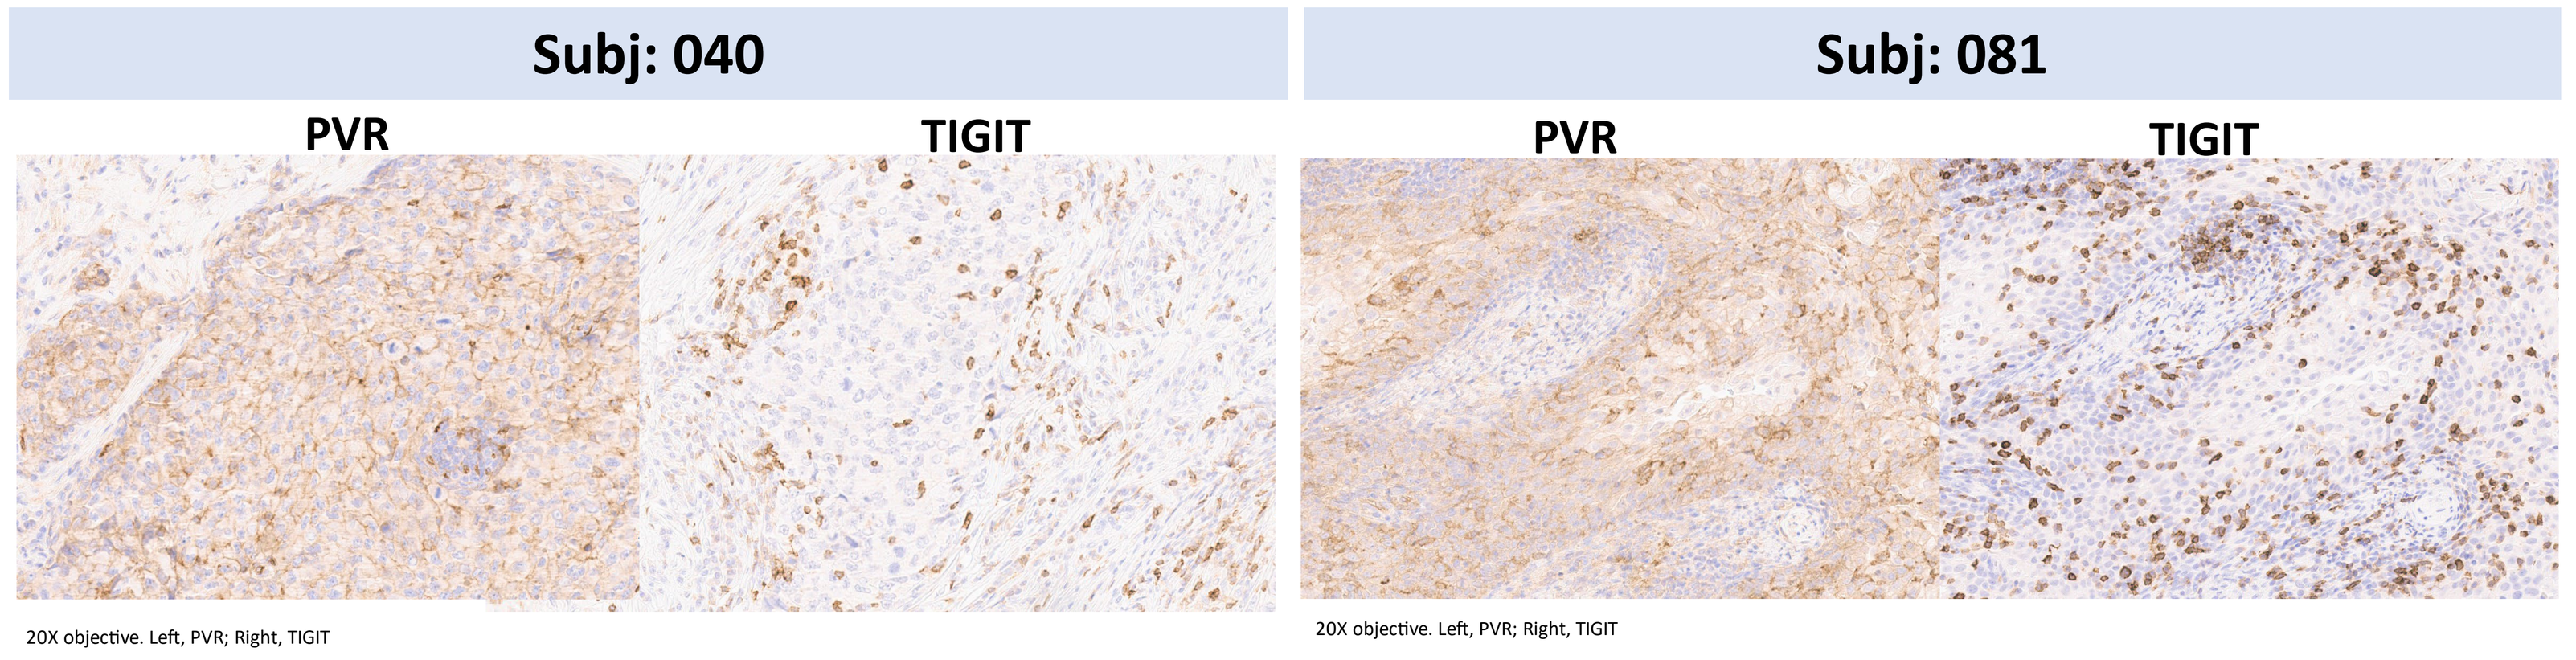
Subj: 040
PVR
TIGIT
Subj: 081
PVR
TIGIT
20X objective. Left, PVR; Right, TIGIT
20X objective. Left, PVR; Right, TIGIT

Figure 1: PVR and TIGIT expression as detected by c-IHC in two cervical cancer patients with highest levels of biomarker detected is shown.

Biomarker Expression in Patients with Objective Response

Table 1: Summary of PVR & TIGIT biomarker expression in patients with objective response

Subject #	Tumor	Response	TIGIT	PVR	PVR score	PDL1
001	Cervical	cCR	N/A	N/A	N/A	positive
040	Cervical	cCR	70	high	80	positive
081	Cervical	cCR	70	high	95	positive
075	Endometrial	cPR	3	high	90	positive
047	Sarcoma (dediff UPS)	cPR	10	high	75	positive
030	Uveal	cPR	0	N/A	N/A	negative
044	Ovarian	cPR	0	high	75	negative
024	Ovarian	uPR	2	high	90	negative
078	Ovarian	uPR	20	high	100	positive

- 030 is PVR and TIGIT/CD3 positive by mIF
- 044 is TIGIT/CD3 positive by mIF, tissue necrosis on central lab slide
- N/A= FFPE not available at central CLIA lab for testing.

Response =Best percent change from baseline of target lesions; Data cutoff 8-12-2022; CLIA IHC assays run in central lab

- No objective responses observed in PVR low patients
- Subjects with PVR low and TIGIT negative tumors progressed at or before first scan (7 of 7)

Biomarker Expression in Patients with Clinical Benefit

Table 2: TIGIT high tumor expression enriched for subjects with clinical benefit

TIGIT	# of Patients	SD	PR	CR	Clinical Benefit	Objective Response
High*	12	3**	2	2	7 (58%)	4 (33%)
Low	42	10	4	0	14 (33%)	4 (10%)

TIGIT high >=10% CPS

* TIGIT high subjects with clinical benefit are PVR positive, except sarcoma 049 with SD; TIGIT high subject (086) with PD is on-going (~100 day) being treated beyond progression
**084 TIGIT high sarcoma on-going subject (day 115) with SD, -29.5% from baseline of target lesions

Pharmacodynamic Changes in Circulating Biomarkers

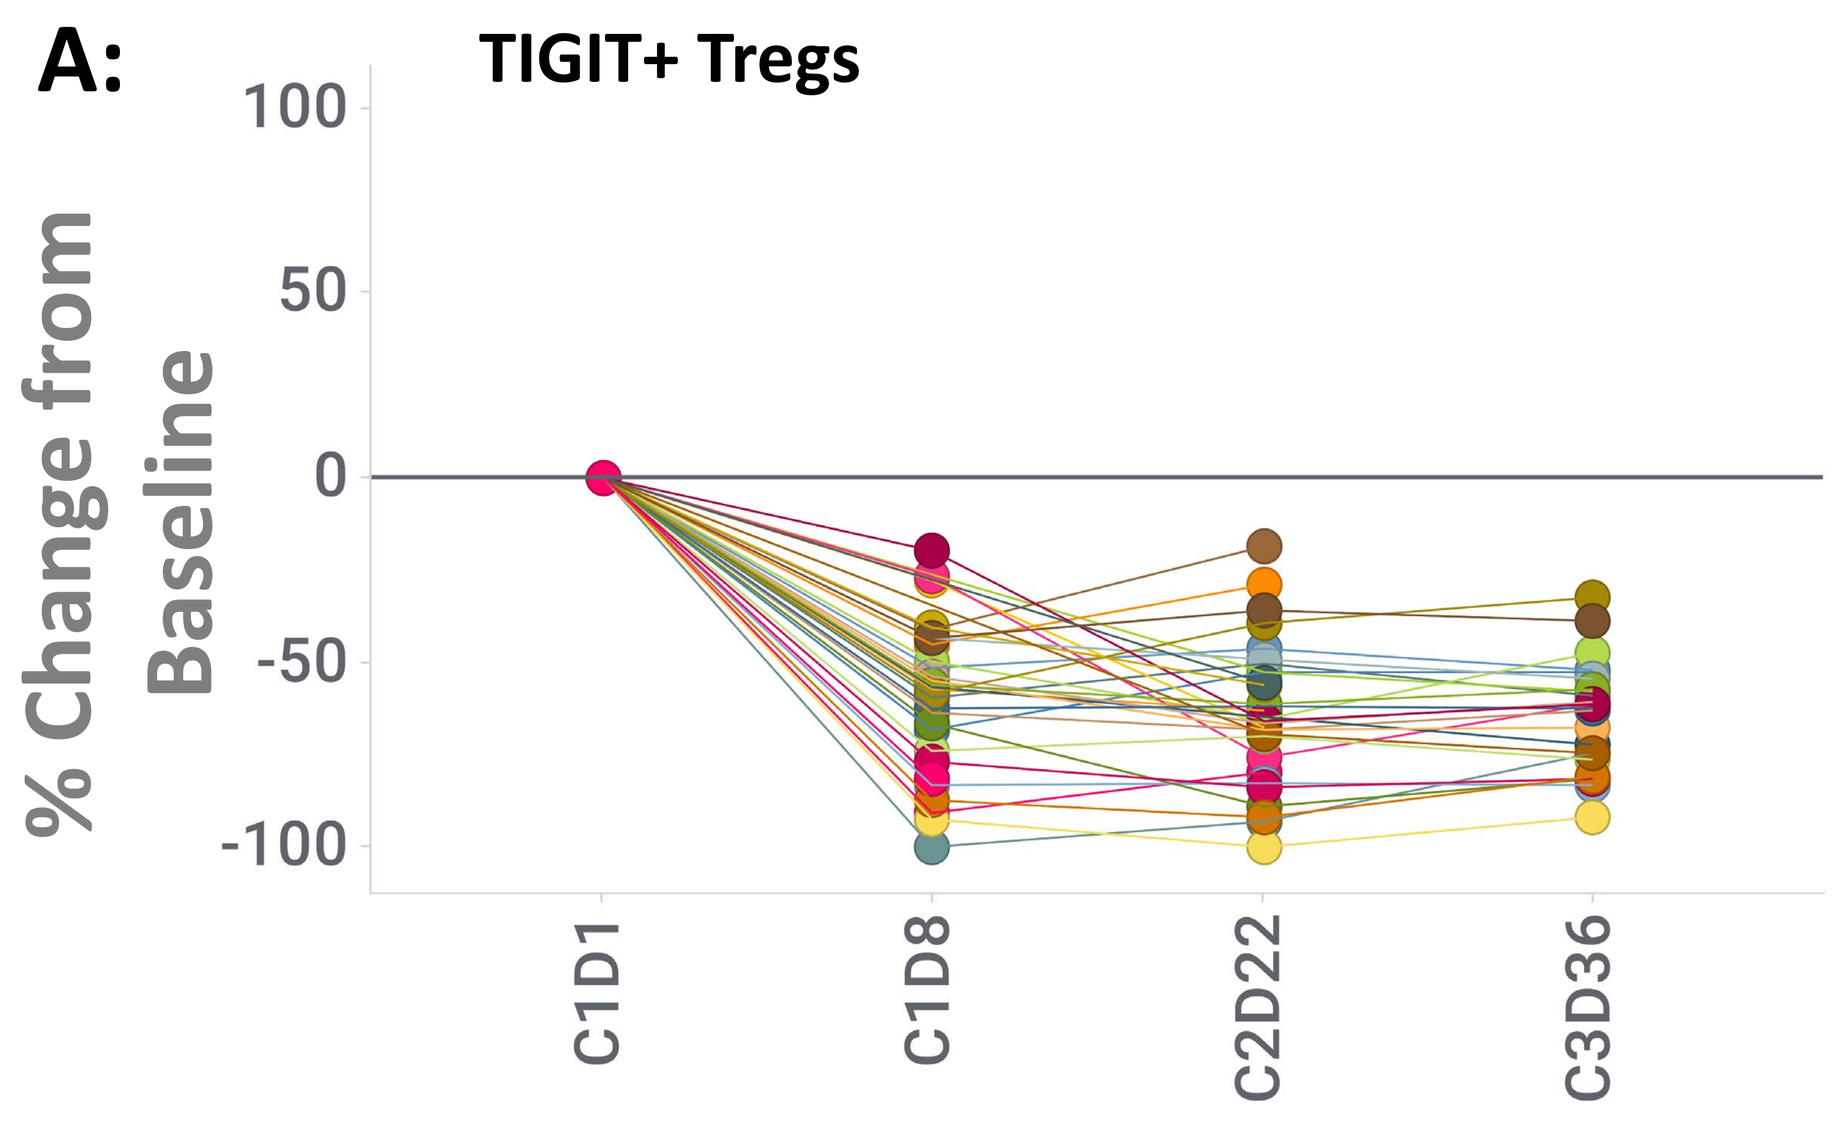
A:
TIGIT+ Tregs
% Change from Baseline
C1D1
C1D8
C2D22
C3D36

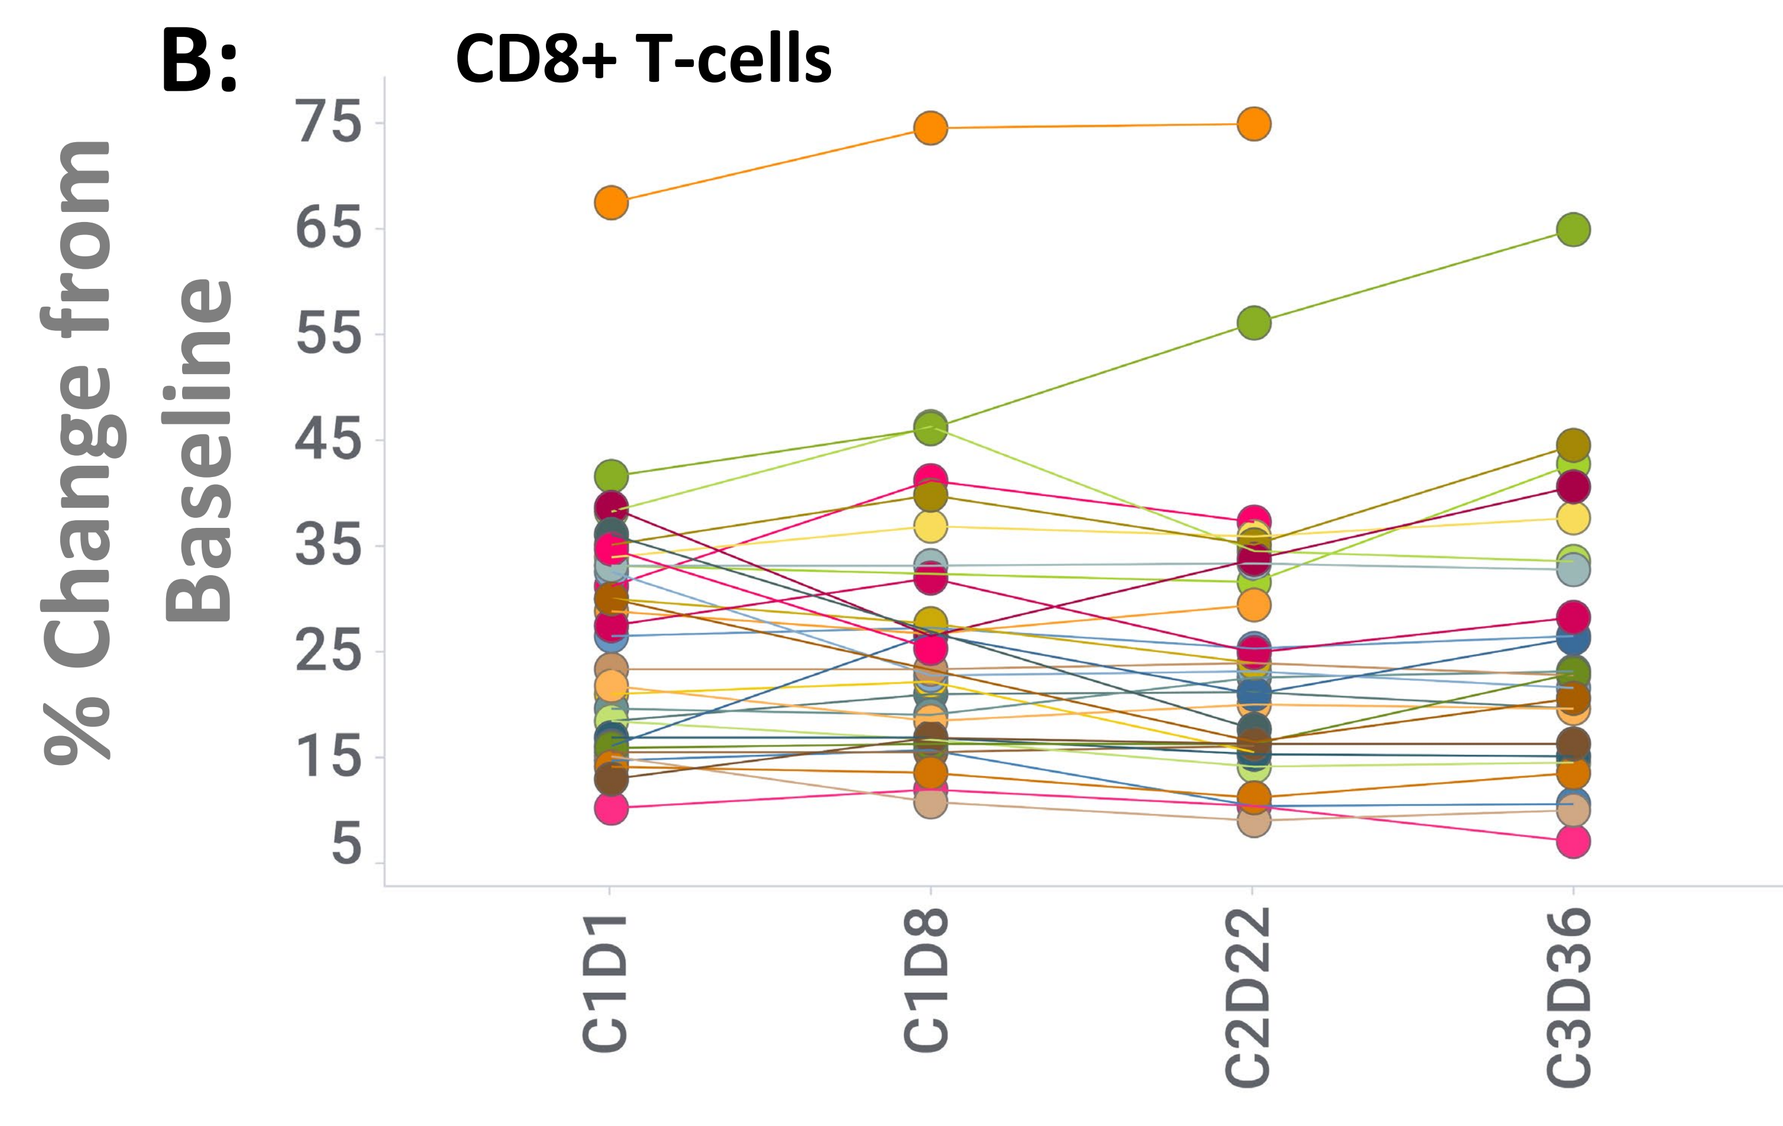
B:
CD8+ T-cells
% Change from Baseline
C1D1
C1D8
C2D22
C3D36

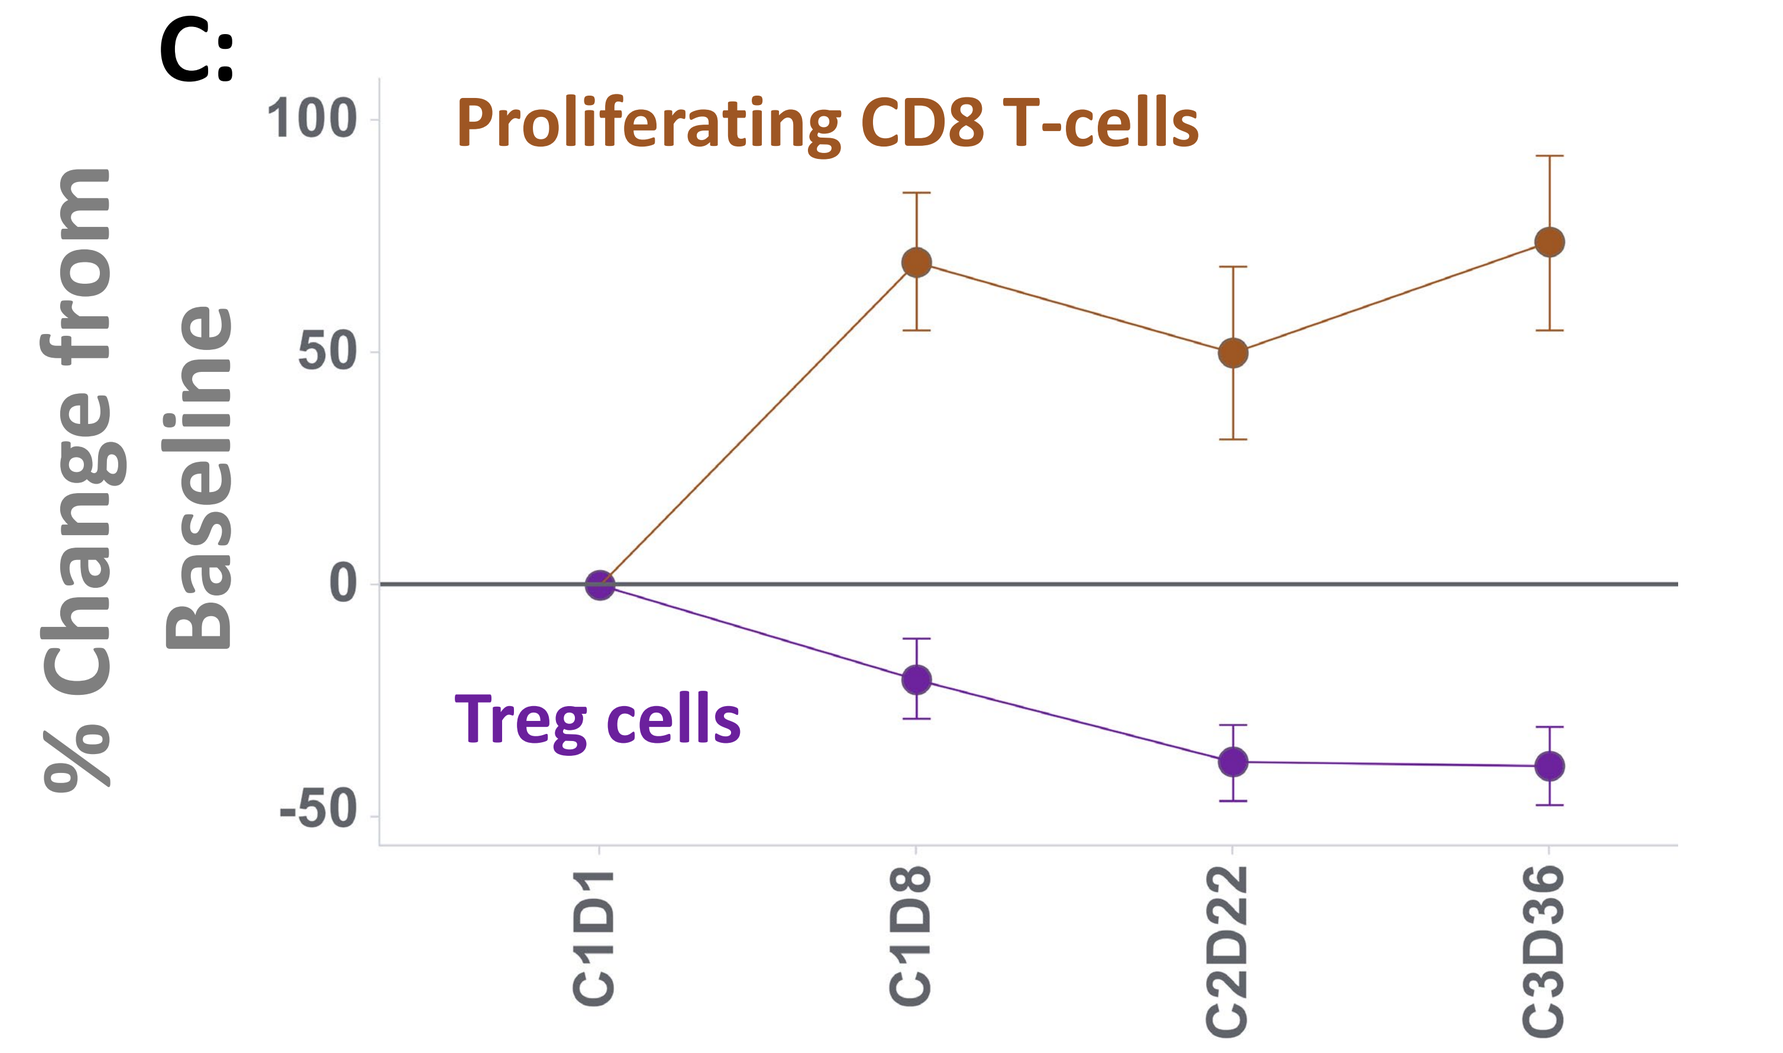
C:
Proliferating CD8 T-cells
Treg cells
% Change from Baseline
C1D1
C1D8
C2D22
C3D36

Figure 2: Robust target engagement is seen through longitudinal changes in PD biomarkers by flow cytometric analysis of PBMCs. (**A**) TIGIT+ Tregs (TIGIT+ CD4+ FoxP3+) cells decrease **(B)** Total CD8+ T-cell frequencies remain stable **(C)** Frequencies of proliferating (Ki-67+) CD8 T-cells increase while total Tregs (CD4+ FoxP3+) decrease. Increases in proliferating **(D)** TIGIT+ CD4+ T-cells and **(E)** NK-cells are seen. **(F)** TPEX[1] (CD8+ CCR7+ PD-1+ TIGIT+) cells decrease while **(G)** increases in TSCM[1] (CD8+ CCR7+ TIGIT- PD-1- CD45RA+) cells were noted. At least 28 subjects were included at baseline.

[1] Galletti et al., Nat Immunol 21, 1552–1562 (2020). https://doi.org/10.1038/s41590-020-0791-5

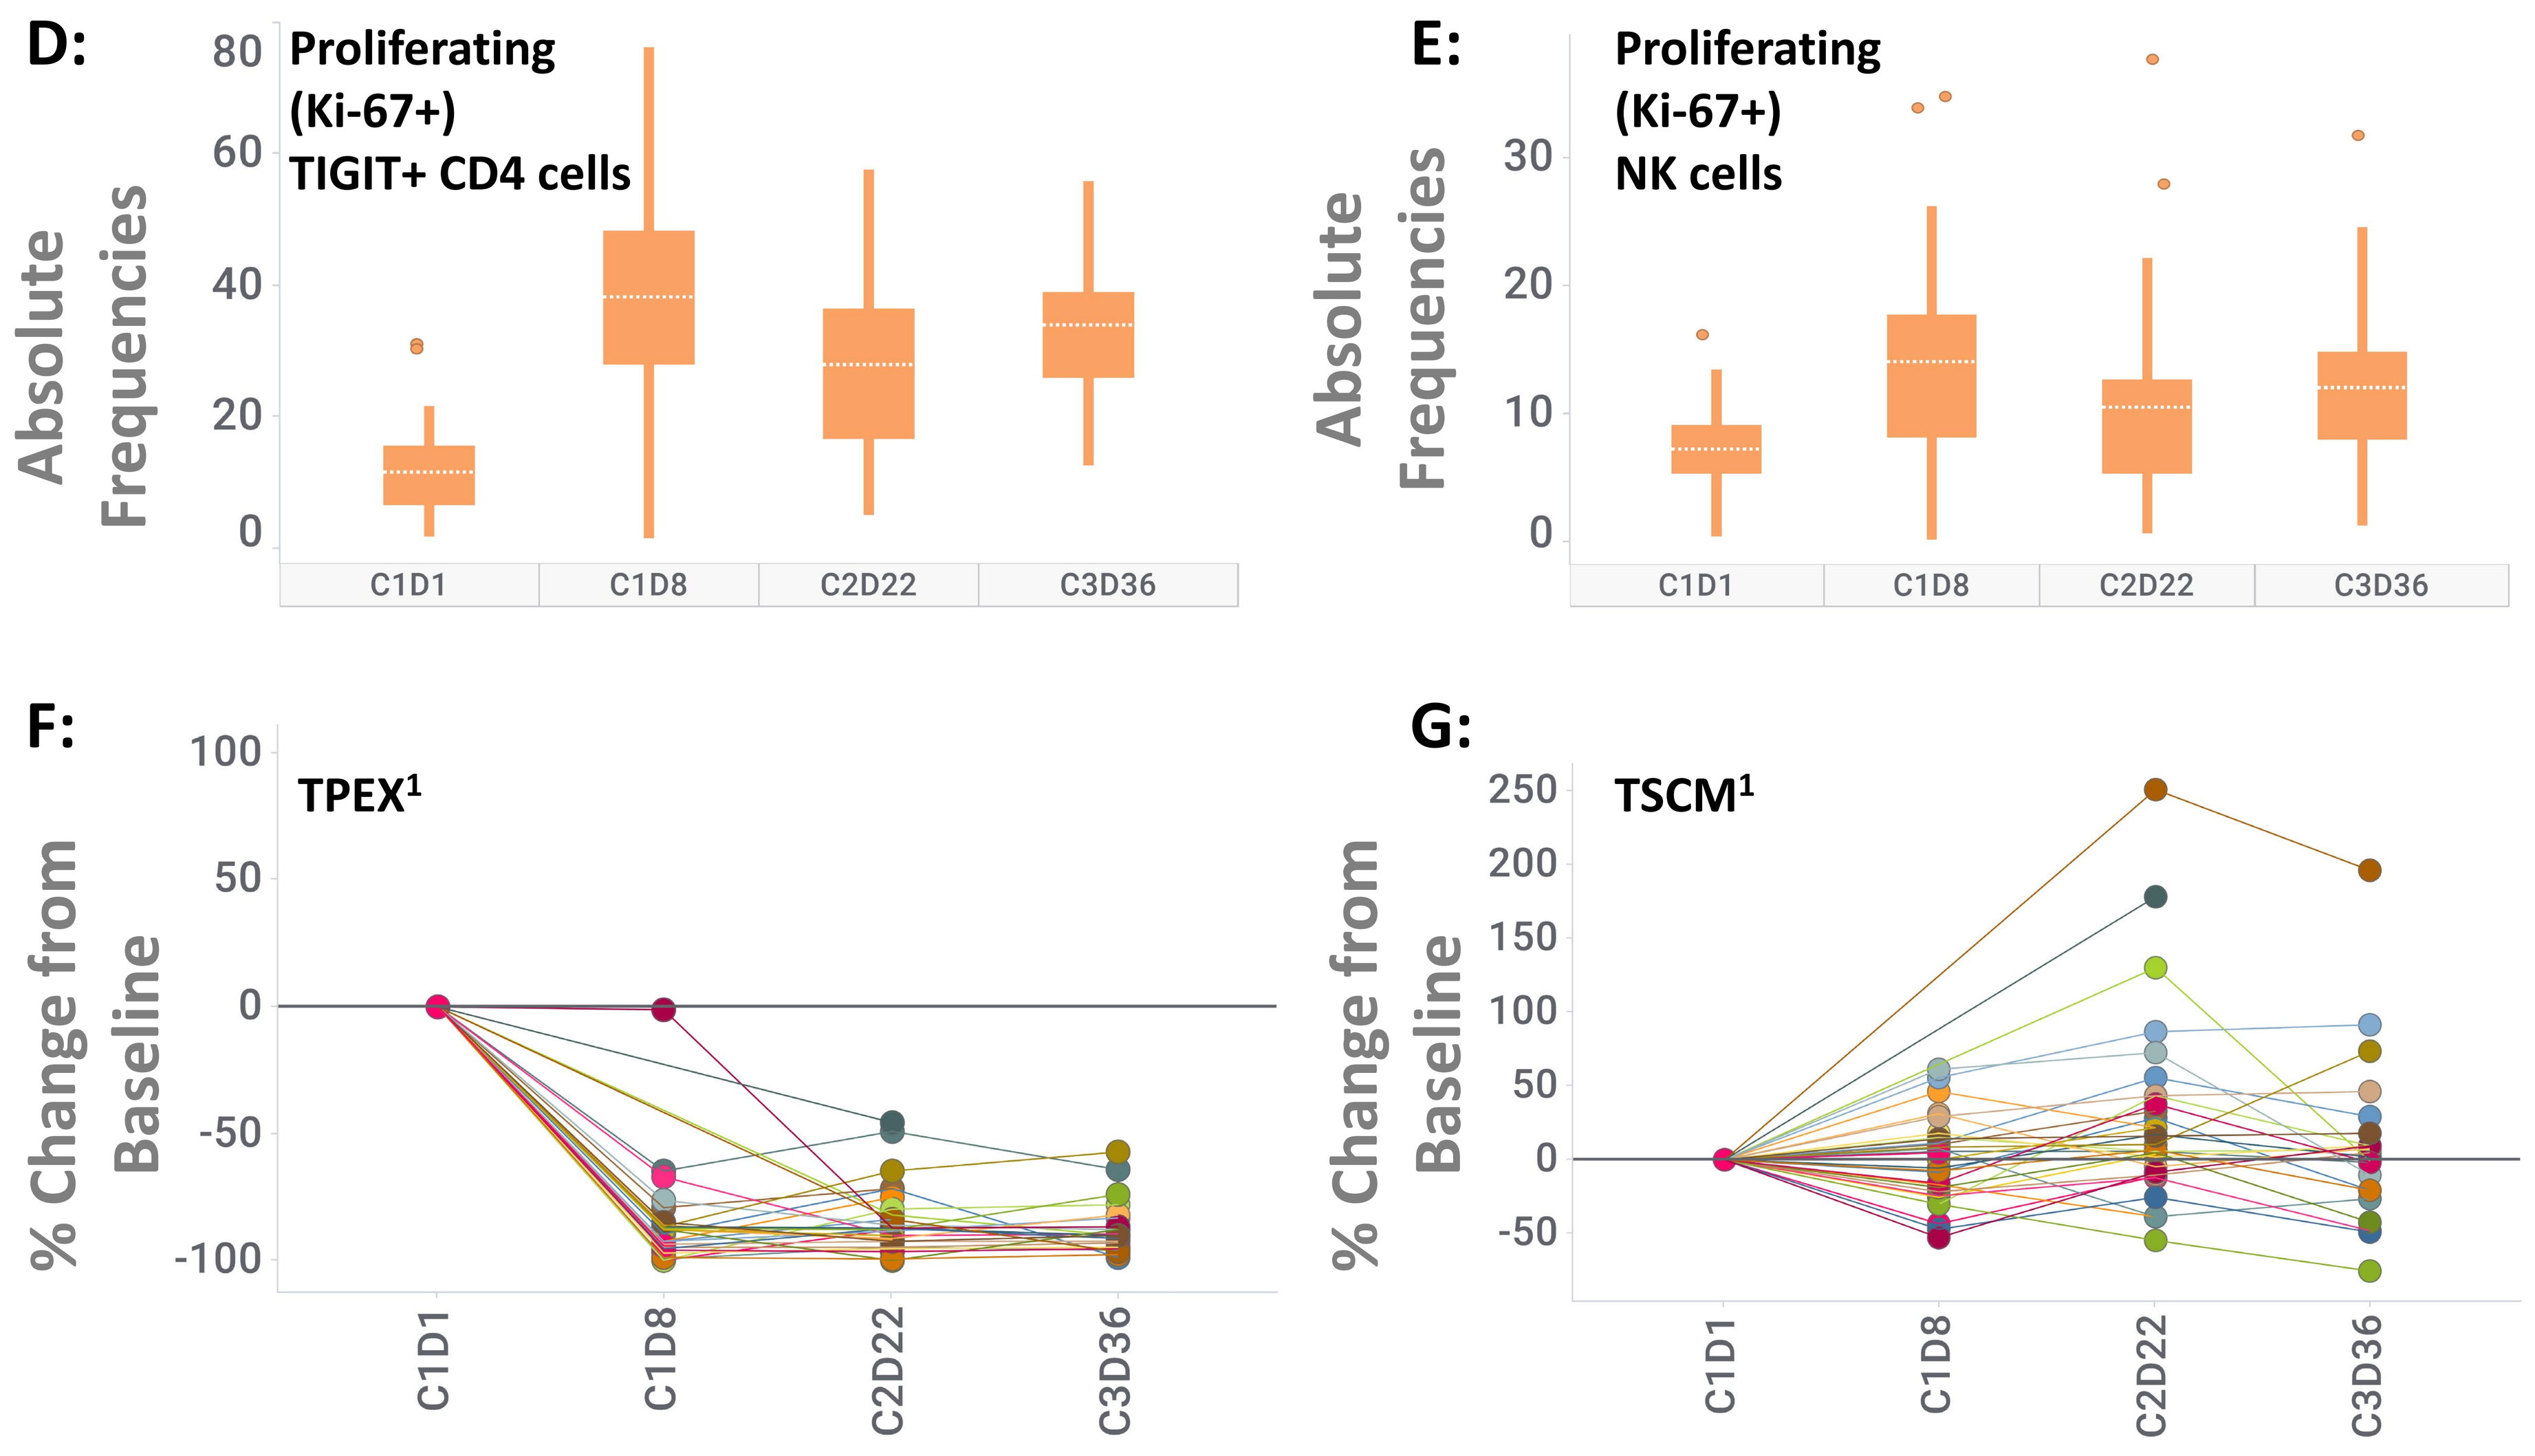
D:
Proliferating (Ki-67+) TIGIT+ CD4 cells
Absolute Frequencies
E:
Proliferating (Ki-67+) NK cells
Absolute Frequencies
F:
TPEX[1]
% Change from Baseline
G:
TSCM[1]
% Change from Baseline
C1D1
C1D8
C2D22
C3D36

Exploratory Gene Signatures in Baseline Tissues and Response

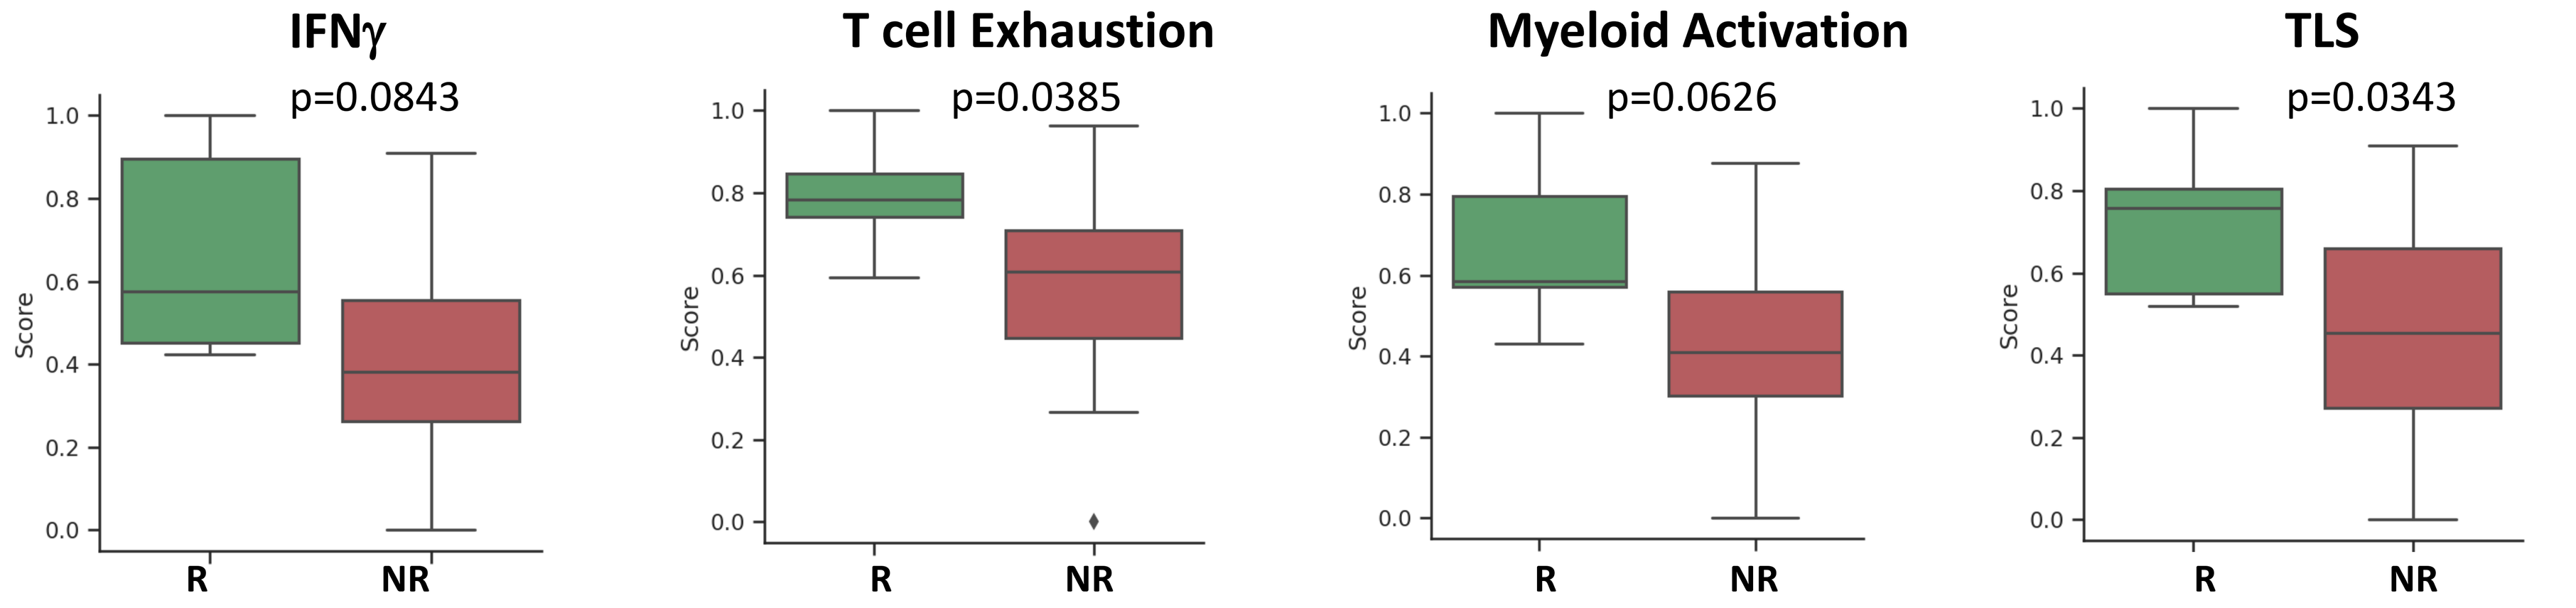
IFNγ
p=0.0843
T cell Exhaustion
p=0.0385
Myeloid Activation
p=0.0626
TLS
p=0.0343
Score
R
NR

Figure 3: Baseline differences in Immune related gene signatures determined by mRNAseq of FFPE tissues were noted between objective responders (CR + PR) and non-responders (SD + PD) corresponding to IFN-γ[1], T-cell Exhaustion[2] Myeloid activation[3] and Tertiary Lymphoid Structures (TLS)[4].

[1]IFNγ=10 gene: J Clin Invest. 2017 Aug 1; 127(8): 2930–2940.
[2]T cell Exhaustion=87 genes_down: Nature. 2015 July 30; 523(7562): 612–616.
[3]Myeloid activation=3 genes: Cancers 2020, 12(6), 1431.
[4]Tertiary lymphoid structure (TLS)=4 genes: Journal for ImmunoTherapy of Cancer 9, no. Suppl 2 (November 2021): A845–A845.

Response =Best percent change from baseline of target lesions; Data cutoff 8-12-2022. n=5 (R)responders (CR/PR); n=33 (N) non-responders (SD/PD)

Changes in Circulating Biomarkers and Clinical Benefit

Figure 4: Changes in biomarkers associated with clinical benefit. **(A)** Greater increases in IP-10, a myeloid cytokine, were noted in patients' plasma with clinical benefit (CR+PR+SD) by ELISA. **(B)** Activated CD4 Effector Memory (EM) cells showed a trend towards increased IFN-γ, IL-2, & TNF-α cytokine production by flow cytometry in patients with clinical benefit. **(C)** Decreases in ctDNA at 5-6 weeks post-treatment determined by Guardant OMNI™ correlated with objective responses. n=14 (A) and n=29 (B) subjects at baseline.

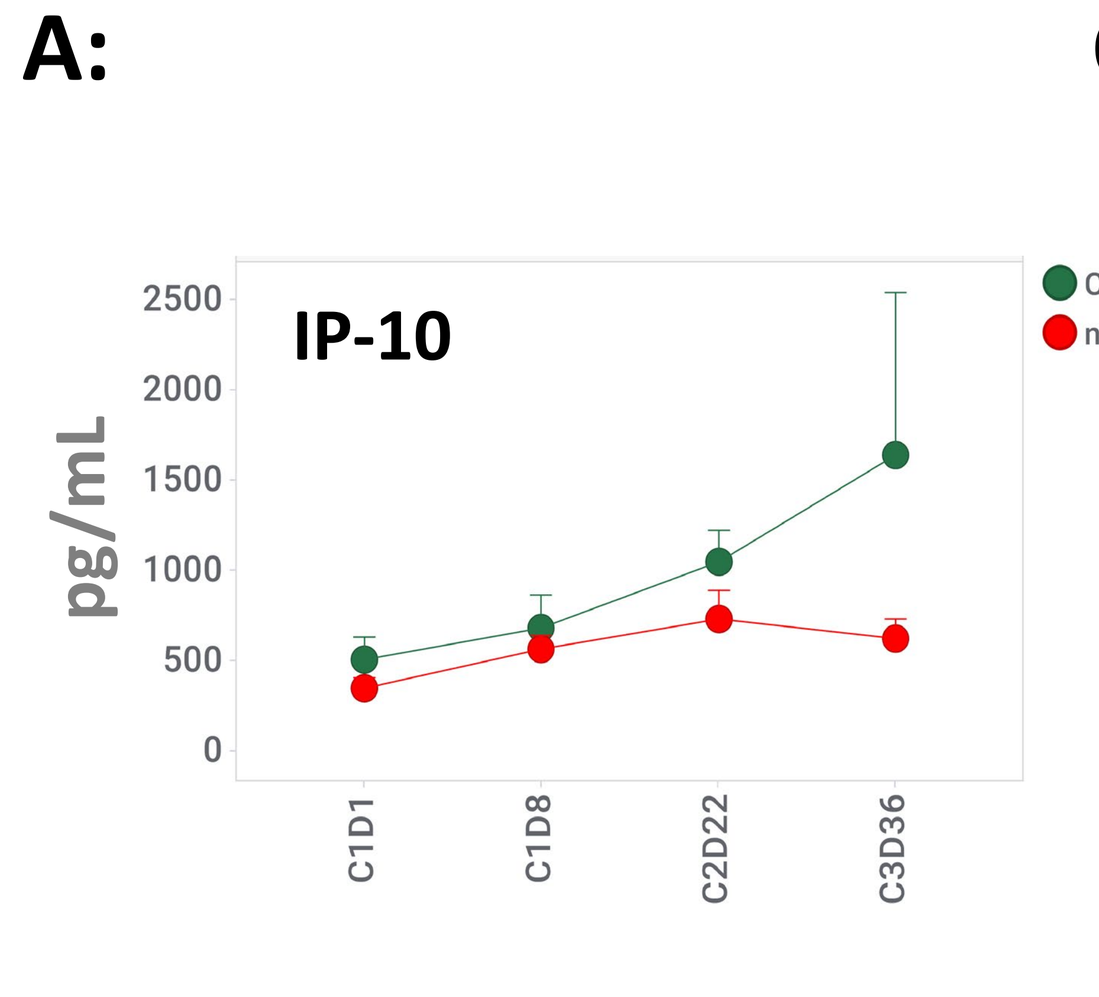
A:
IP-10
pg/mL
CB
no CB
C1D1
C1D8
C2D22
C3D36

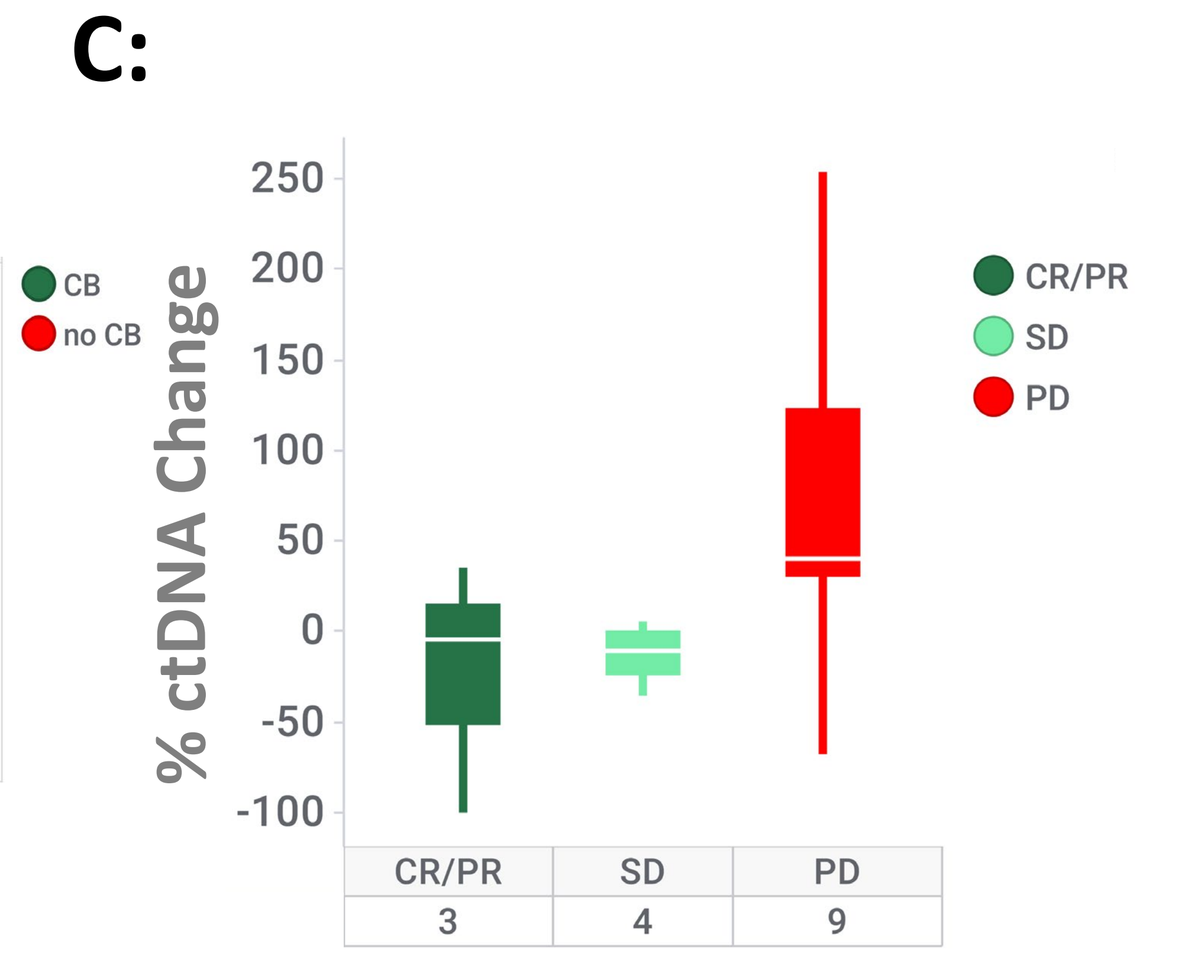
C:
% ctDNA Change
CR/PR
SD
PD
3
4
9

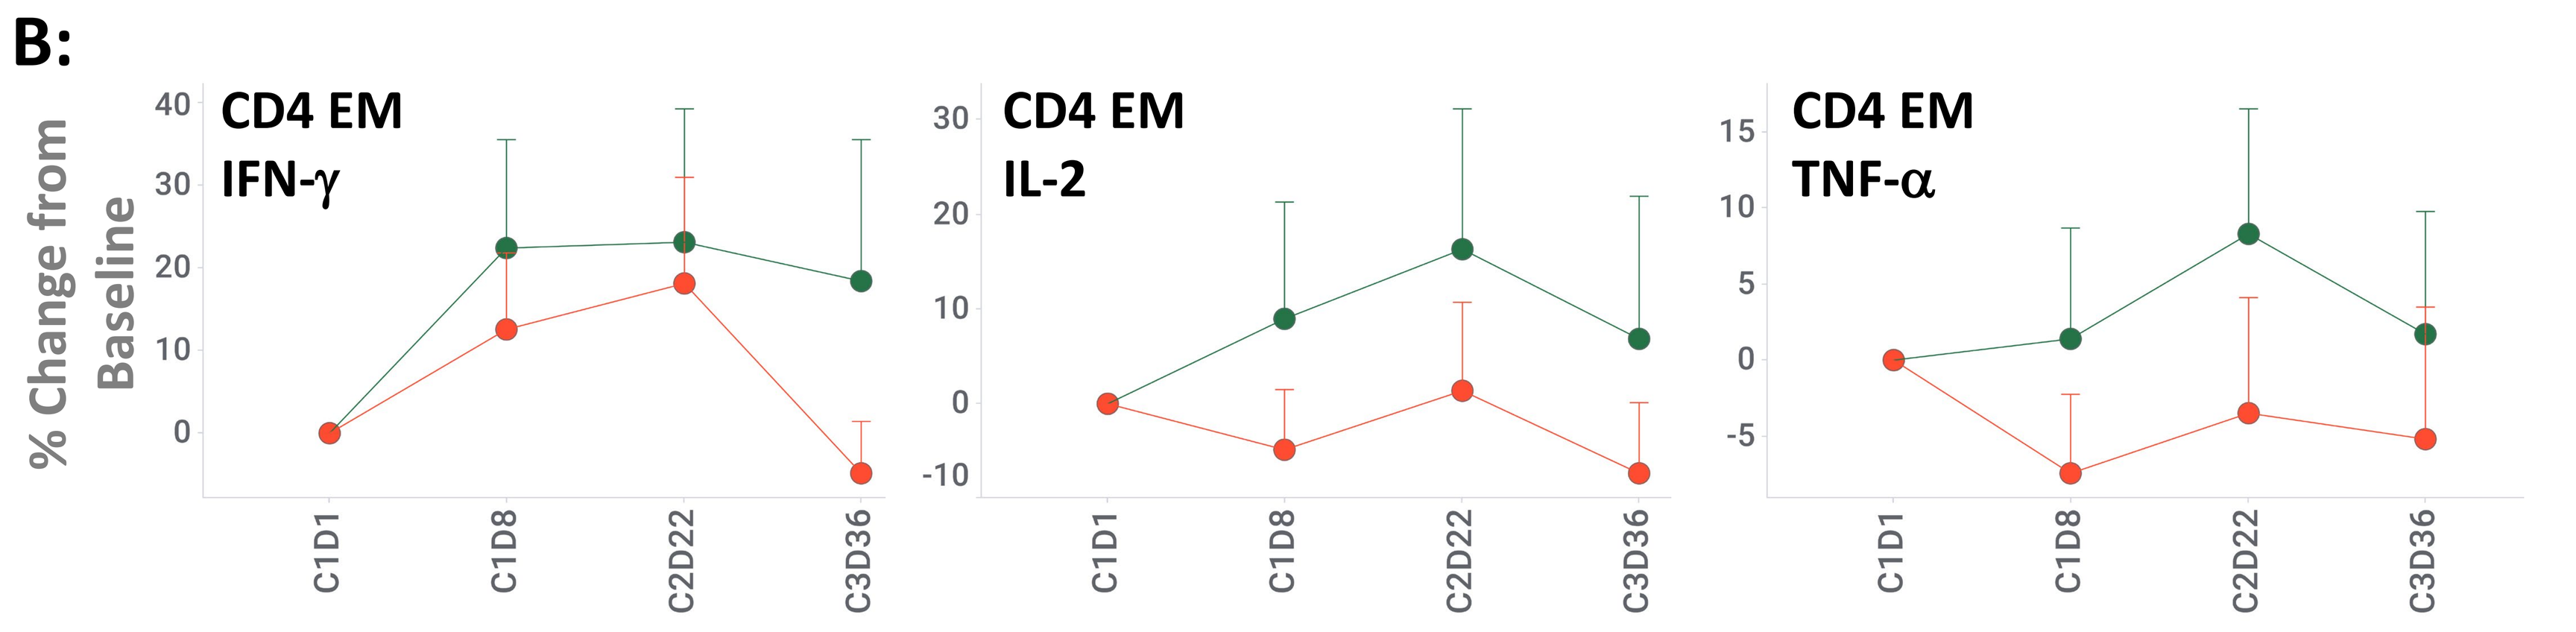
B:
% Change from Baseline
CD4 EM IFN-γ
CD4 EM IL-2
CD4 EM TNF-α
C1D1
C1D8
C2D22
C3D36

Tissue Biomarker Expression at Baseline Associates with Tumor Shrinkage

Figure 5: Tissue biomarker expression at baseline shows association with clinical benefit. **(A)** PVR expression determined by multiplex Immuno-fluorescence demonstrated robust PVR expression in tumor regions. **(B)** Subjects with tumor shrinkage or no growth from baseline expressed PVR at higher levels compared to those subjects that had increases in tumor growth.

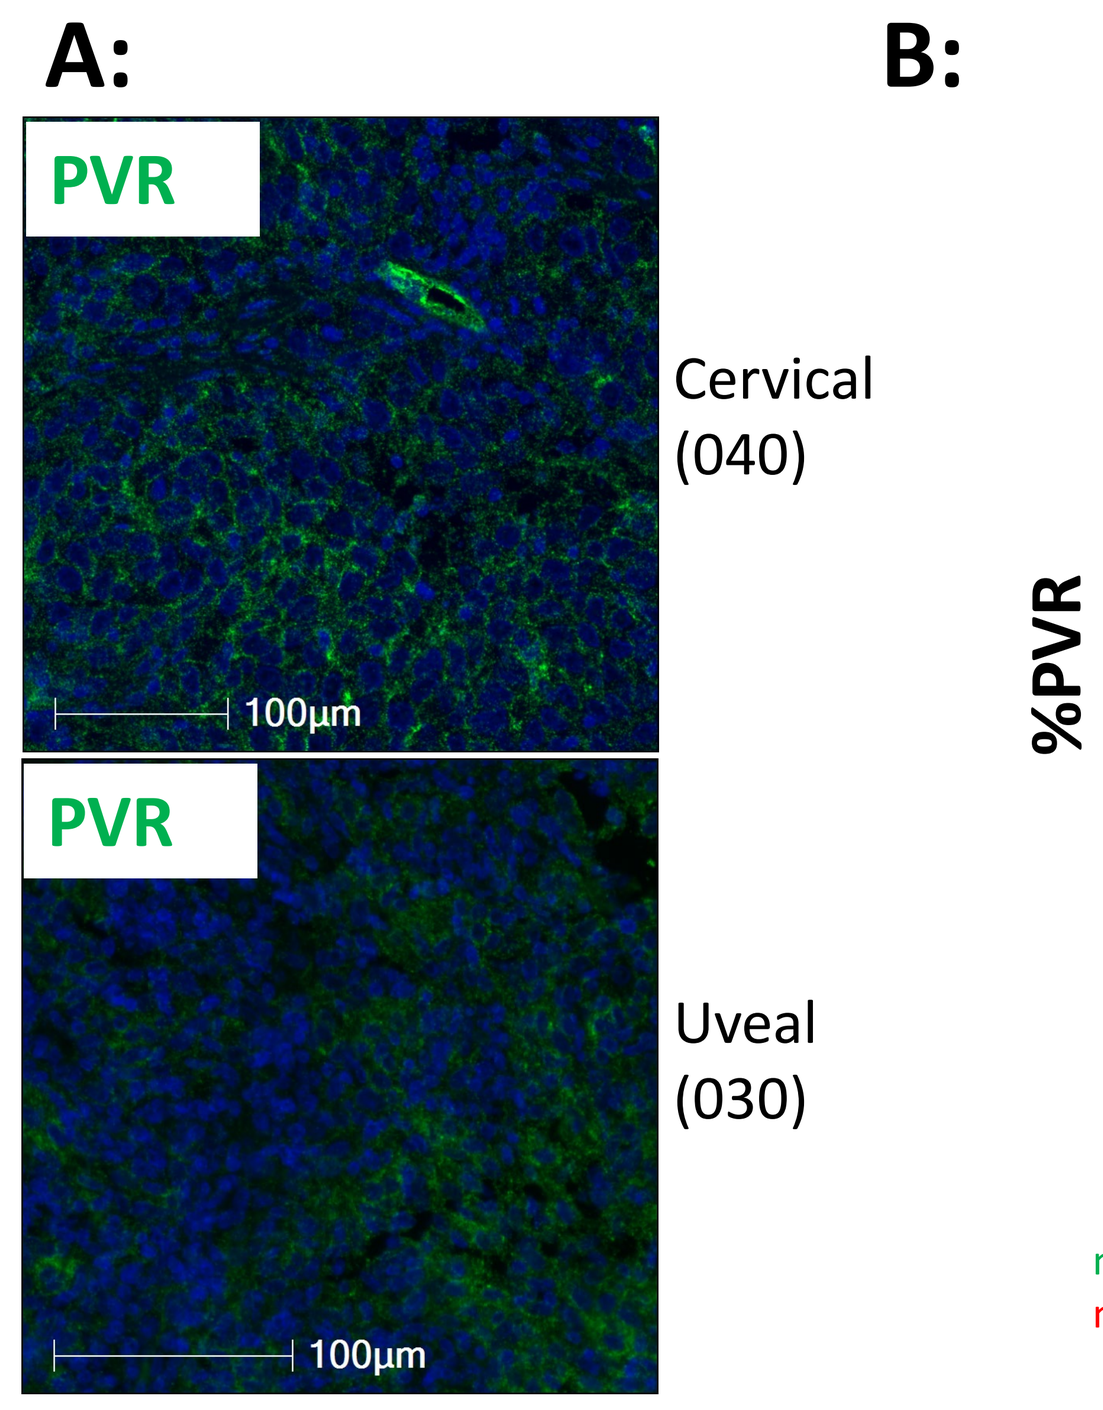
A:
PVR
100µm
Cervical (040)
PVR
100µm
Uveal (030)

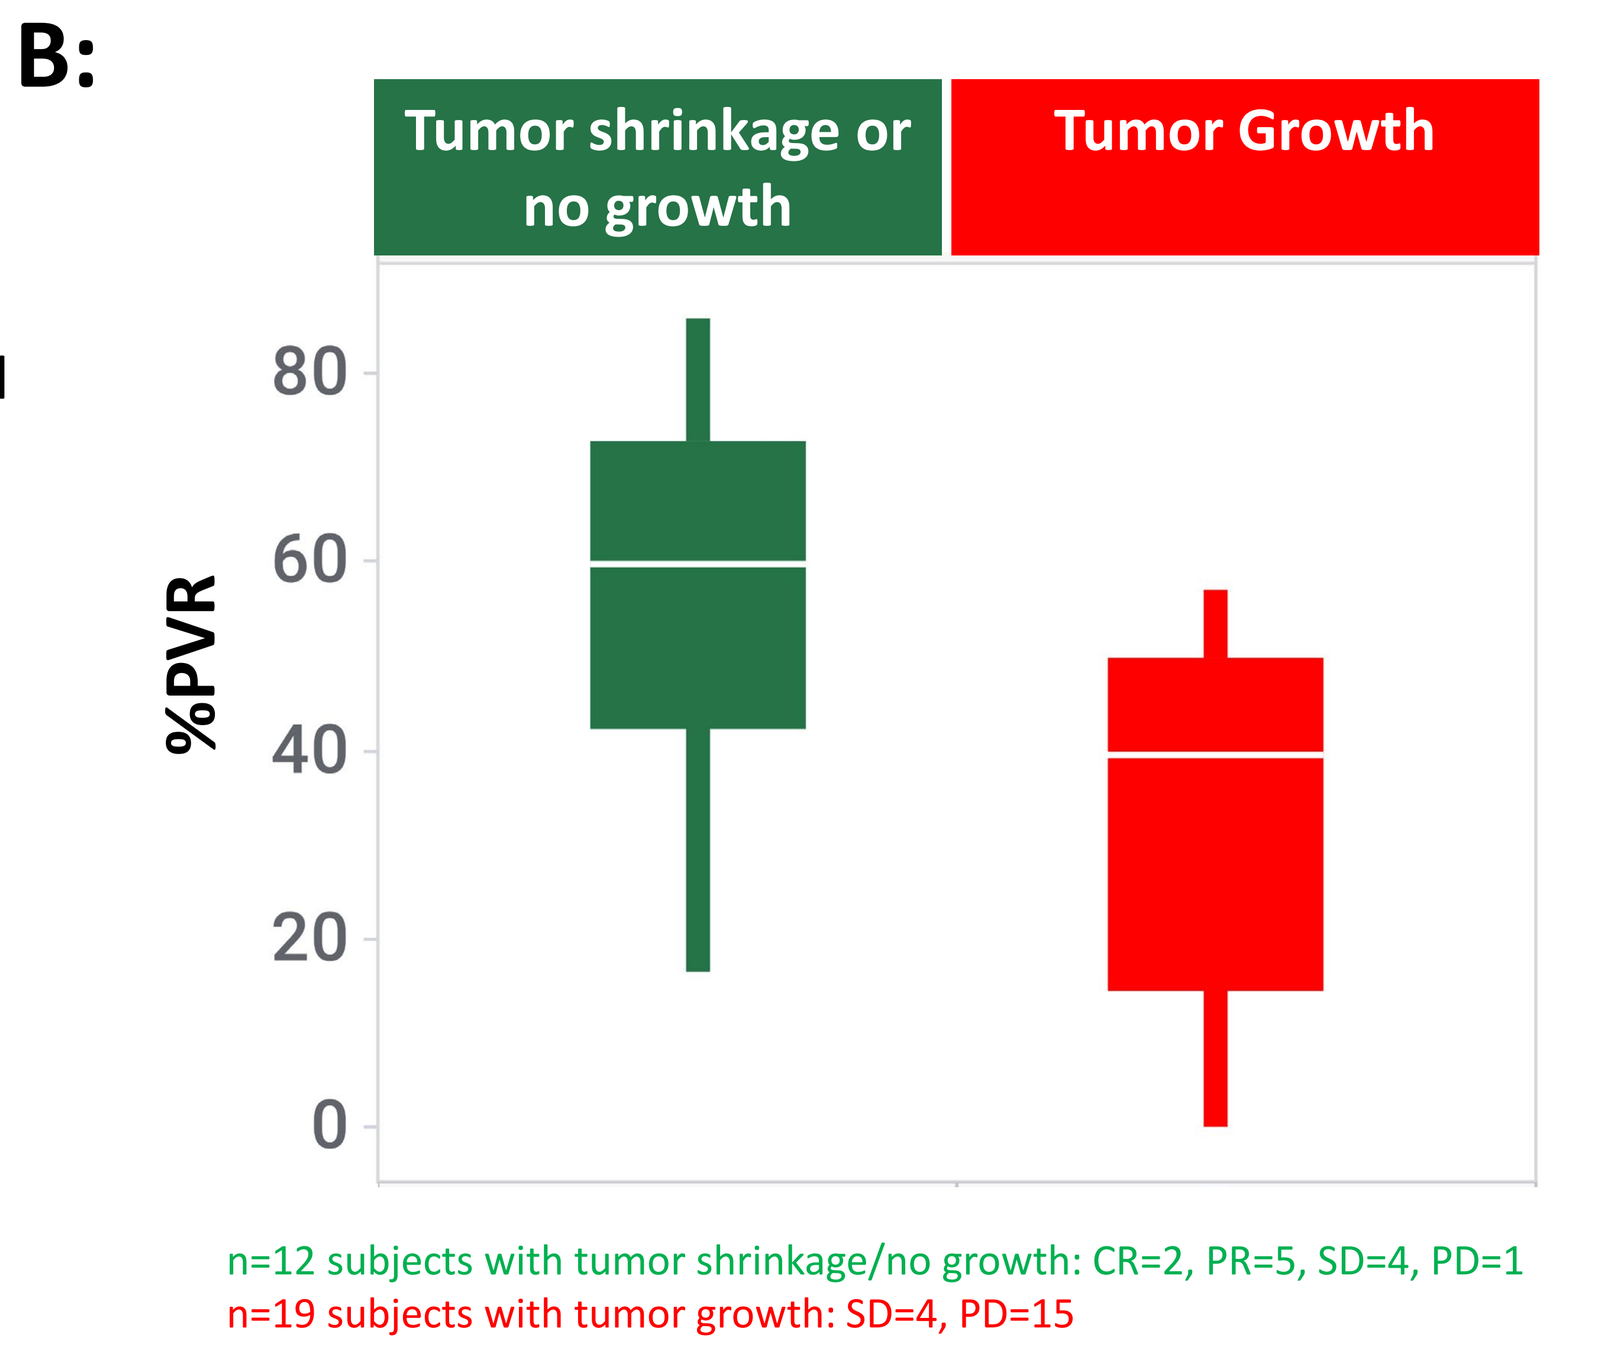
B:
Tumor shrinkage or no growth
Tumor Growth
%PVR

n=12 subjects with tumor shrinkage/no growth: CR=2, PR=5, SD=4, PD=1
n=19 subjects with tumor growth: SD=4, PD=15

Co-Expression of CD226+/CD8+ by Multiplex Immunofluorescence

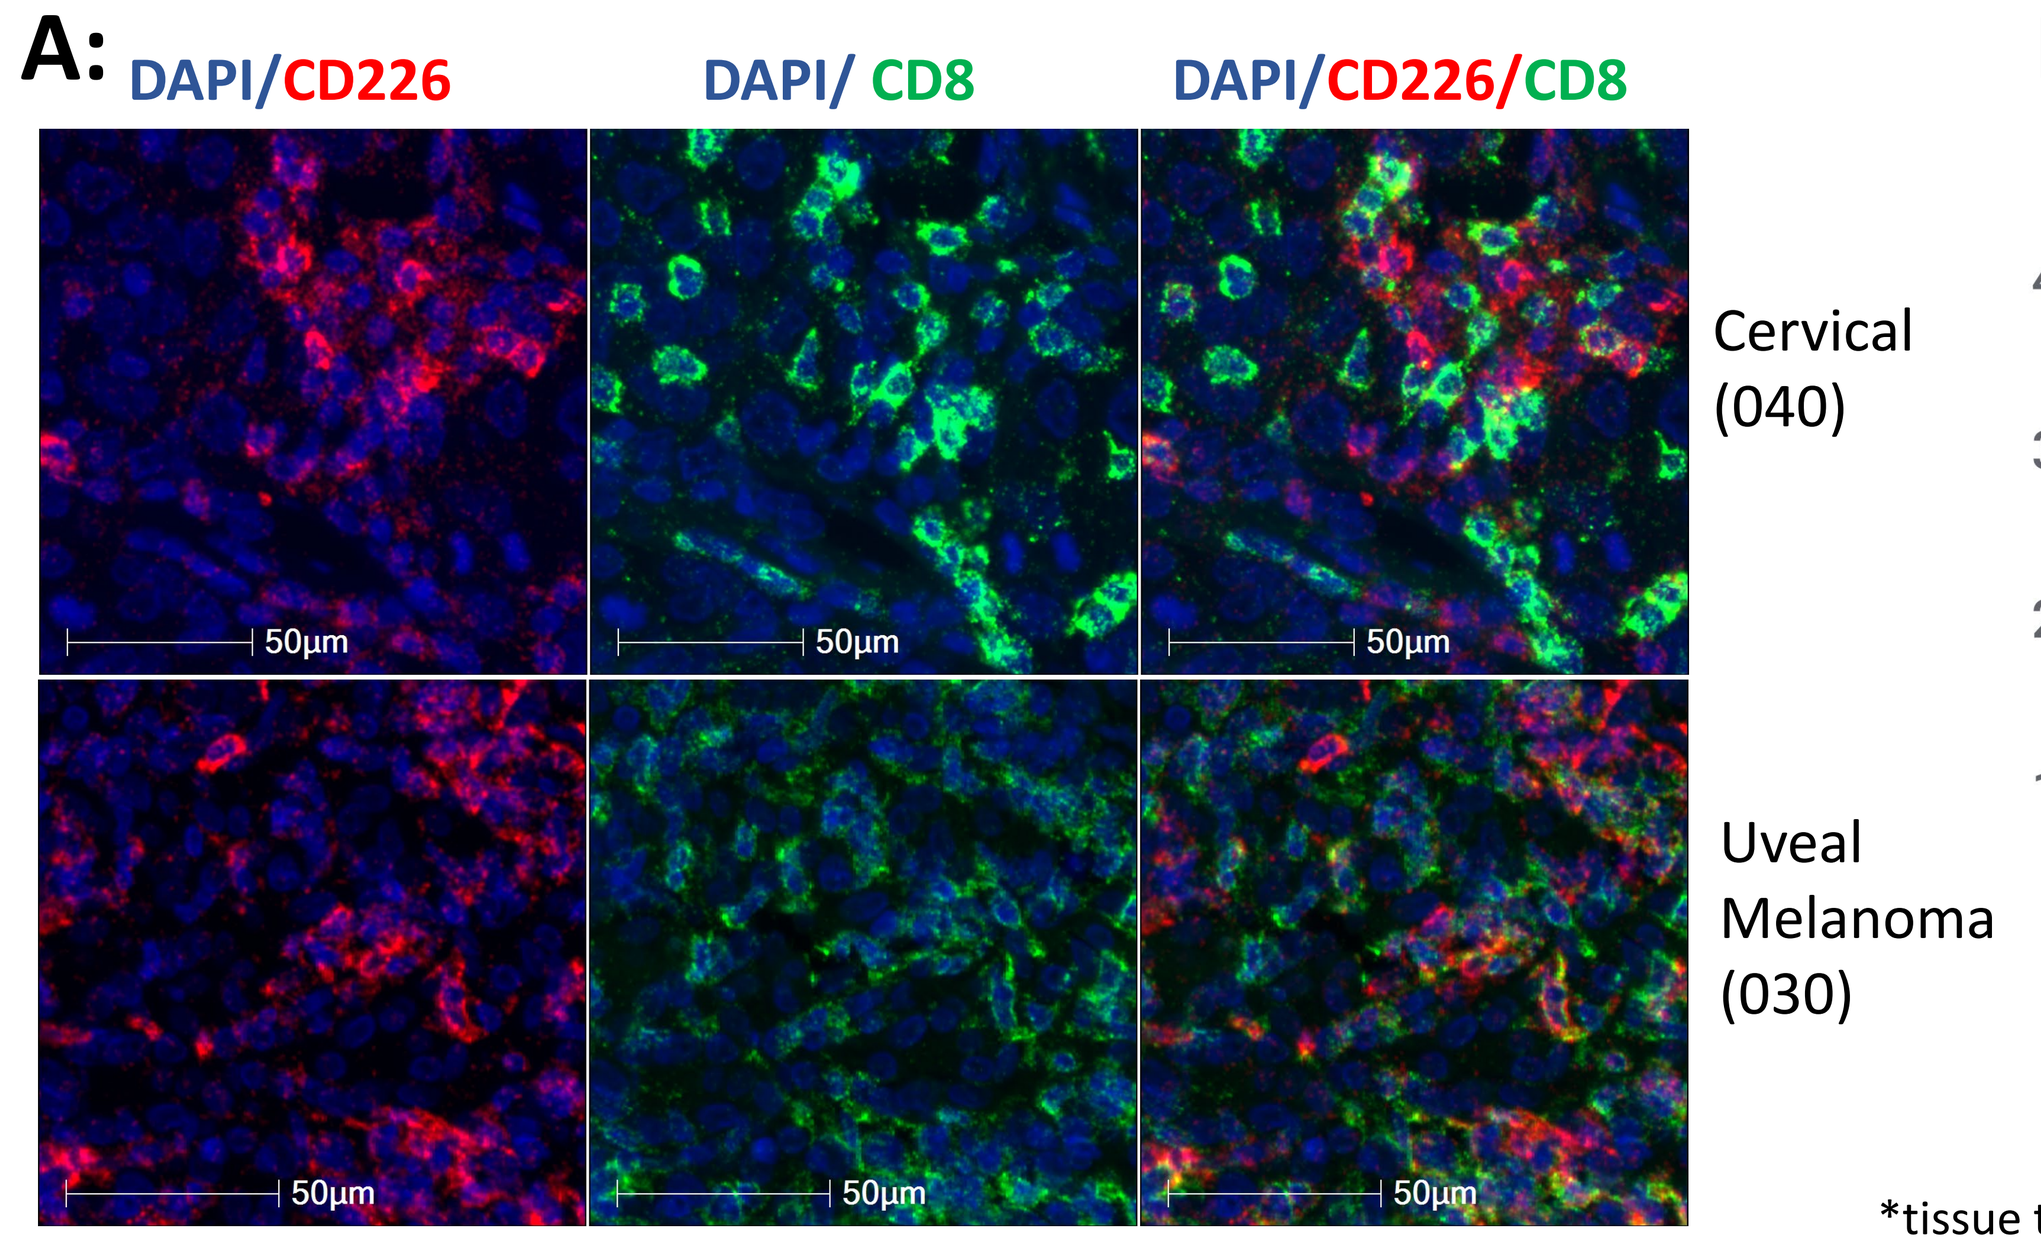
A:
DAPI/CD226
DAPI/ CD8
DAPI/CD226/CD8
50µm
Cervical (040)
Uveal Melanoma (030)

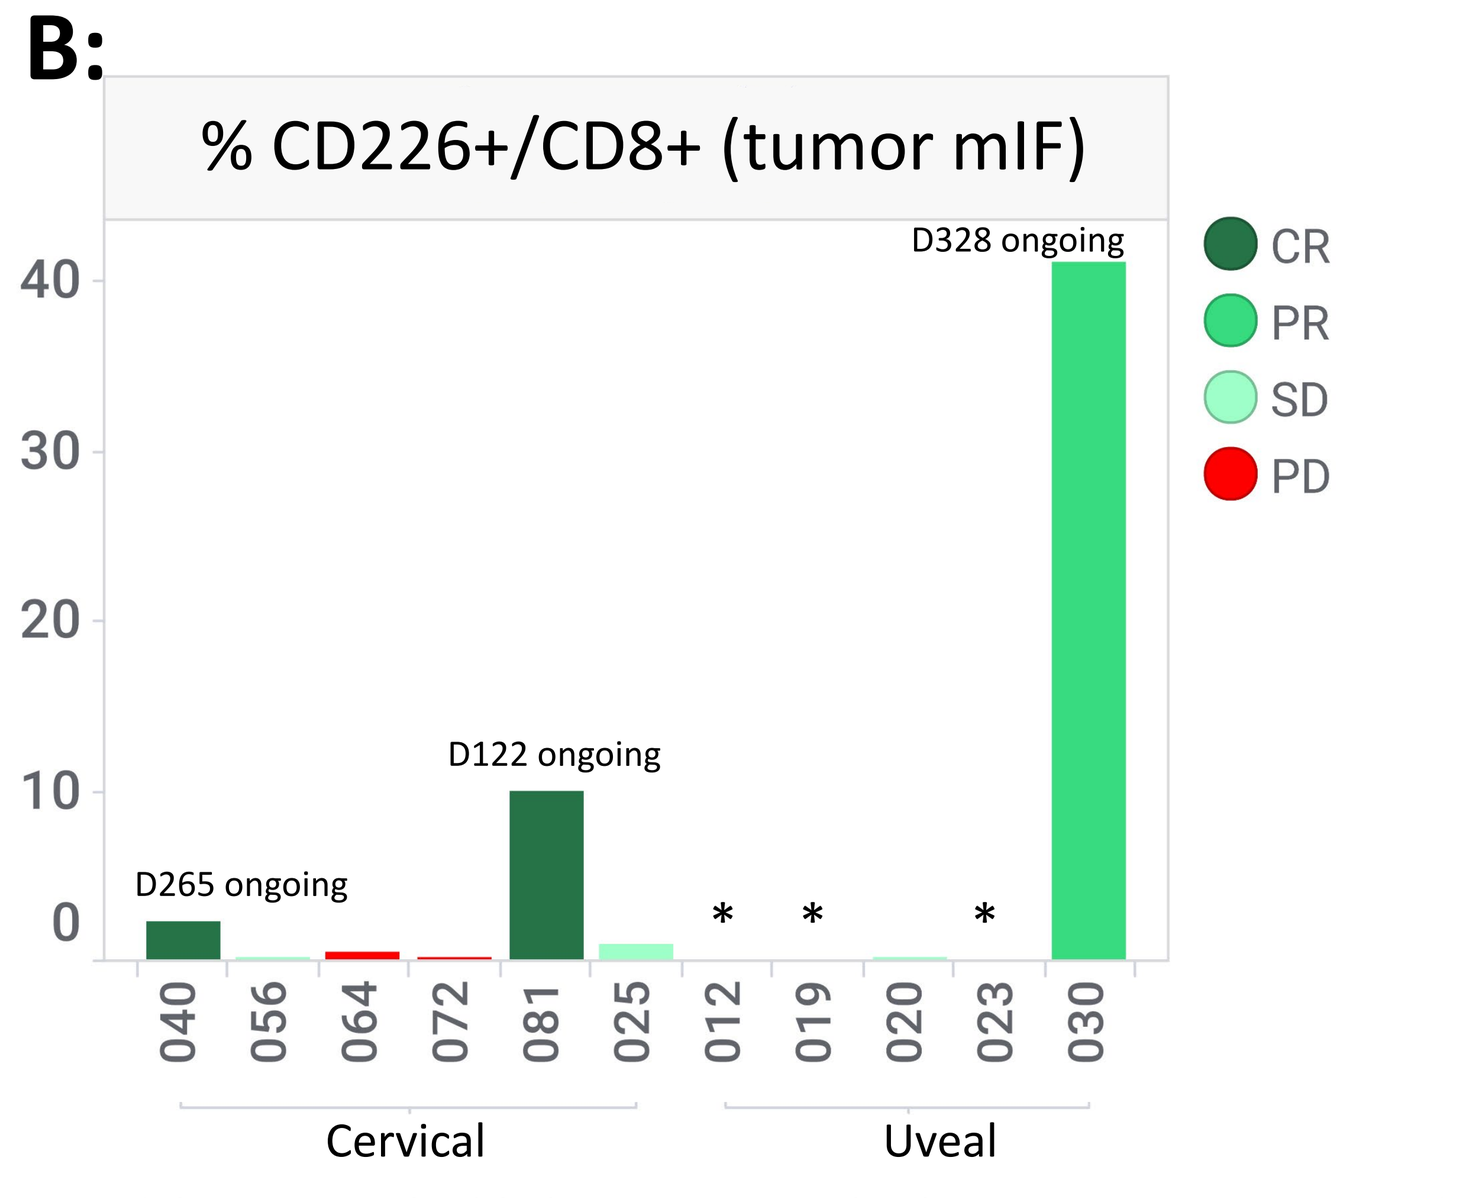
B:
% CD226+/CD8+ (tumor mIF)
CR
PR
SD
PD
D328 ongoing
D122 ongoing
D265 ongoing
040
056
064
072
081
025
012
019
020
023
030
Cervical
Uveal

*tissue tested, but not evaluable by mIF; cervical subject #001 was not tested due to no tumor tissue. #030 tumor tissue shows high immune infiltration

Figure 6: **(A)** Baseline tissue biomarker analysis showed co-expression of CD226 and CD8 in tumor regions of cervical and uveal melanoma subjects with objective response. **(B)** Percent of cells positive for CD226 and CD8 were highest in 030 followed by 081 and 040 respectively.

Conclusions

- Robust target engagement observed
- ctDNA reductions correlation with clinical response
- Data support further evaluation of PVR, TIGIT and CD226 as potential biomarkers of enrichment to etigilimab plus anti-PD1

Disclosure: Ghanashyam Sarikonda (shyam.sarikonda@mereobiopharma.com) is an employee of Mereo Biopharma. Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.